Antitumor Activity of CUDC-907, a Dual PI3K and HDAC Inhibitor in Hematological Cancer Models

Rudi Bao, Cheng-Jung Lai, Dagong Wang, Hui Qu, Ling Yin, Brian Zifcak, Guang-Xin Xu, Steven DellaRocca, Mylissa Borek, Ruzanna Atoyan, Maria Samson, Anna Ma, Jing Wang, Hai-Xaio Zhai, Xiong Cai, Carmen Pepicelli, Changgeng Qian, Maurizio Voi

Curis Inc., 4 Maguire Road, Lexington, MA 02421

2012 AACR Poster # 3744

Rationale for Inhibiting Both PI3K and HDAC in Human Hematological Cancer Xenografts

- Both PI3k and HDAC are validated targets for hematological cancers
- Inhibiting both targets simultaneously with a single agent as opposed to using a combination of agents inhibiting each target individually may be able to improve antitumor activity
- A single agent approach may address mismatched pharmacological properties, potential drug-drug interactions, improve patient compliance, reduce costs associated with two branded agents, and overcome higher technical and regulatory hurdles when using two targeted agents in combination

CUDC-907 Is an Orally Bio-available Small Molecule Inhibitor against Both PI3K and HDAC

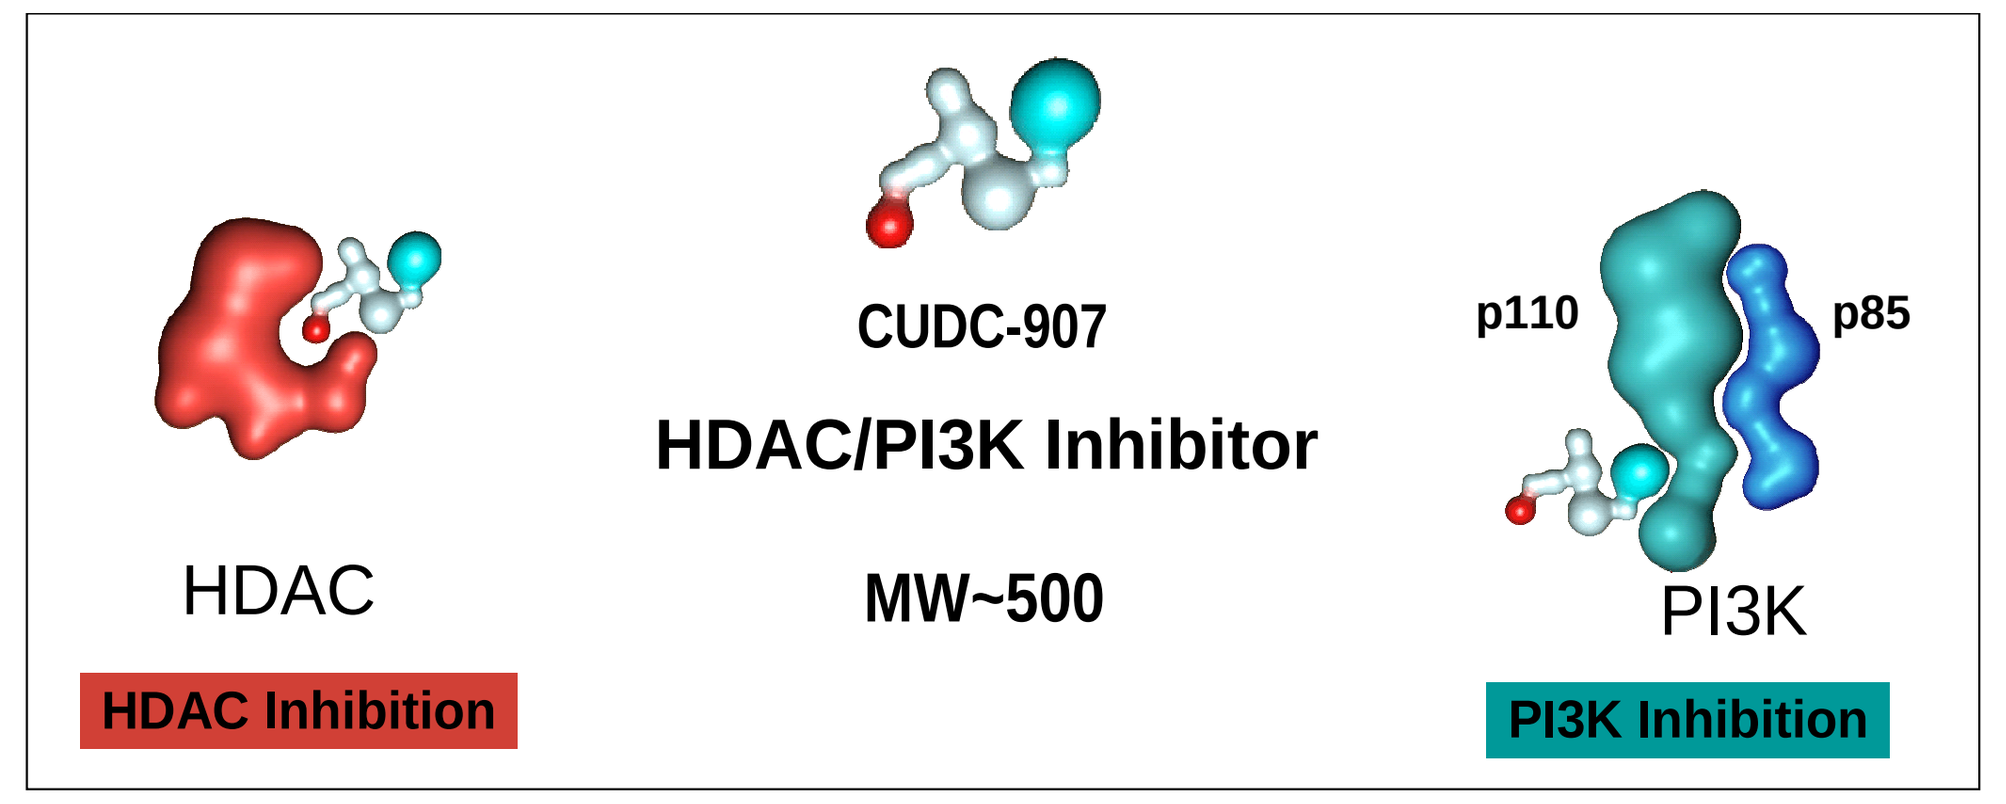
HDAC
HDAC Inhibition
CUDC-907
HDAC/PI3K Inhibitor
MW~500
p110
p85
PI3K
PI3K Inhibition

•Enzymatic activity against PI3K isoforms (IC50, nM)

Compound	PI3Kα	PI3Kβ	PI3Kγ	PI3Kδ	PI3KαH1047R	PI3Kα E545K
CUDC-907	19	54	311	39	73	62

•Enzymatic activity against HDAC subtypes (IC50, nM)

Class	I				II						IV
HDACs	HDAC1	HDAC2	HDAC3	HDAC8	HDAC4	HDAC5	HDAC6	HDAC7	HDAC9	HDAC10	HDAC11
CUDC-907	1.7	5.0	1.8	191	409	674	27	426	554	2.8	5.4

CUDC-907 Potently Inhibits Proliferation of Hematological Cancer Cell Lines

IC50 (μM)

Cancer Type	Cell Line	SAHA	GDC-0941	SAHA + GDC-0941	CUDC-907	CAL-101	Known mutations
		IC50 (uM)					
B cell lymphoma	Granta 519 (MCL)	3.02	>20	0.59	0.007	>20	
	DOHH2 (Follicular)	0.57	0.025	0.013	0.001	0.055	p16INK4
	RL (DLBCL)	0.94	1.34	0.15	0.002	>20	p53
	Pfeiffer (DLBCL)	2.73	0.55	0.37	0.004	ND	
	SuDHL4 (DLBCL)	1.15	0.45	0.25	0.003	9.73	
	Daudi (Burkitt's)	1.23	>20	0.89	0.015	>20	p53
	Raji (Burkitt's)	3.56	>20	3.9	0.009	>20	p53
Myeloma	RPMI8226	1.01	>20	0.69	0.002	>20	p53, k-Ras, EGFRT751I
	OPM-2	0.43	0.05	0.11	0.001	ND	PTEN
	ARH77	1.42	14.79	0.83	0.005	ND	p53

CUDC-907 Efficacy in Various Multiple Myeloma Models

OPM2

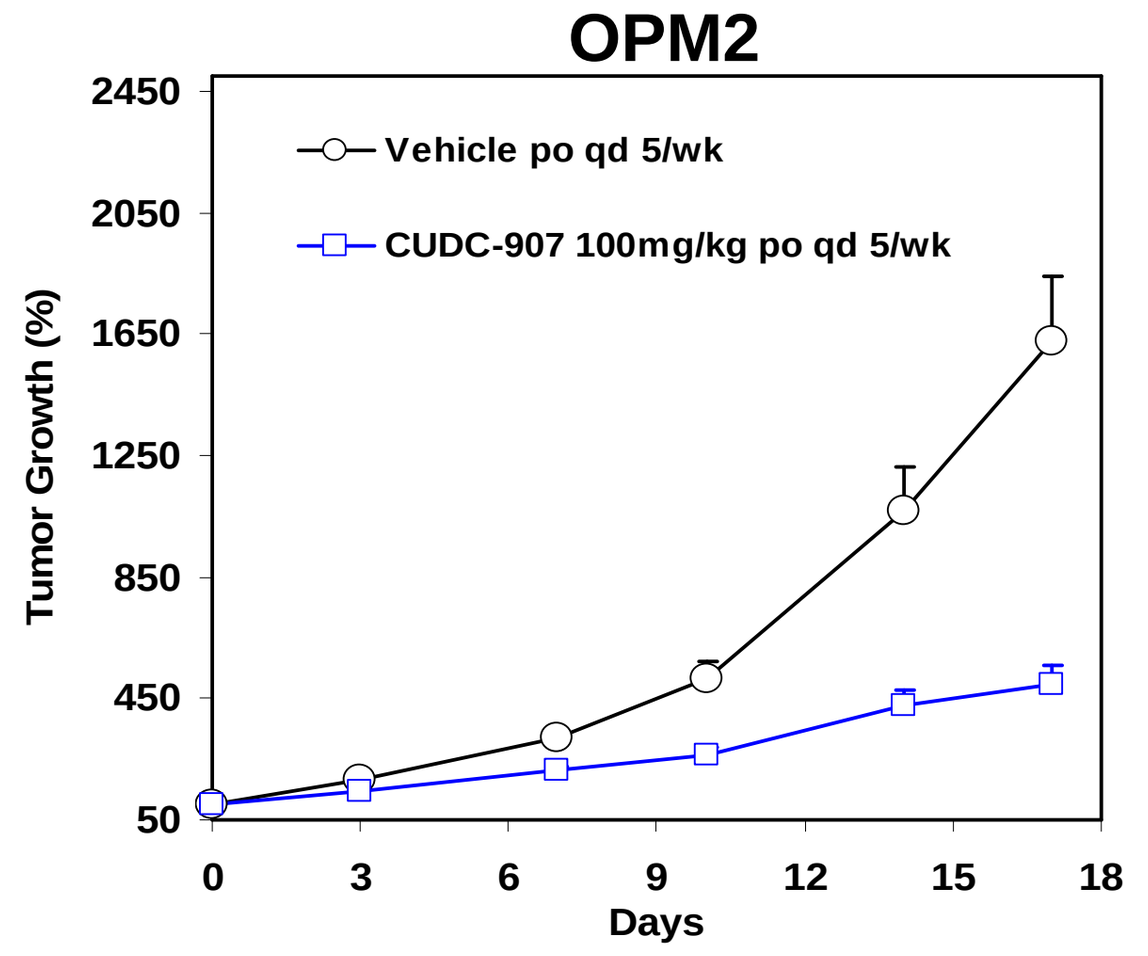
Vehicle po qd 5/wk
CUDC-907 100mg/kg po qd 5/wk
Tumor Growth (%)
Days

MM1S

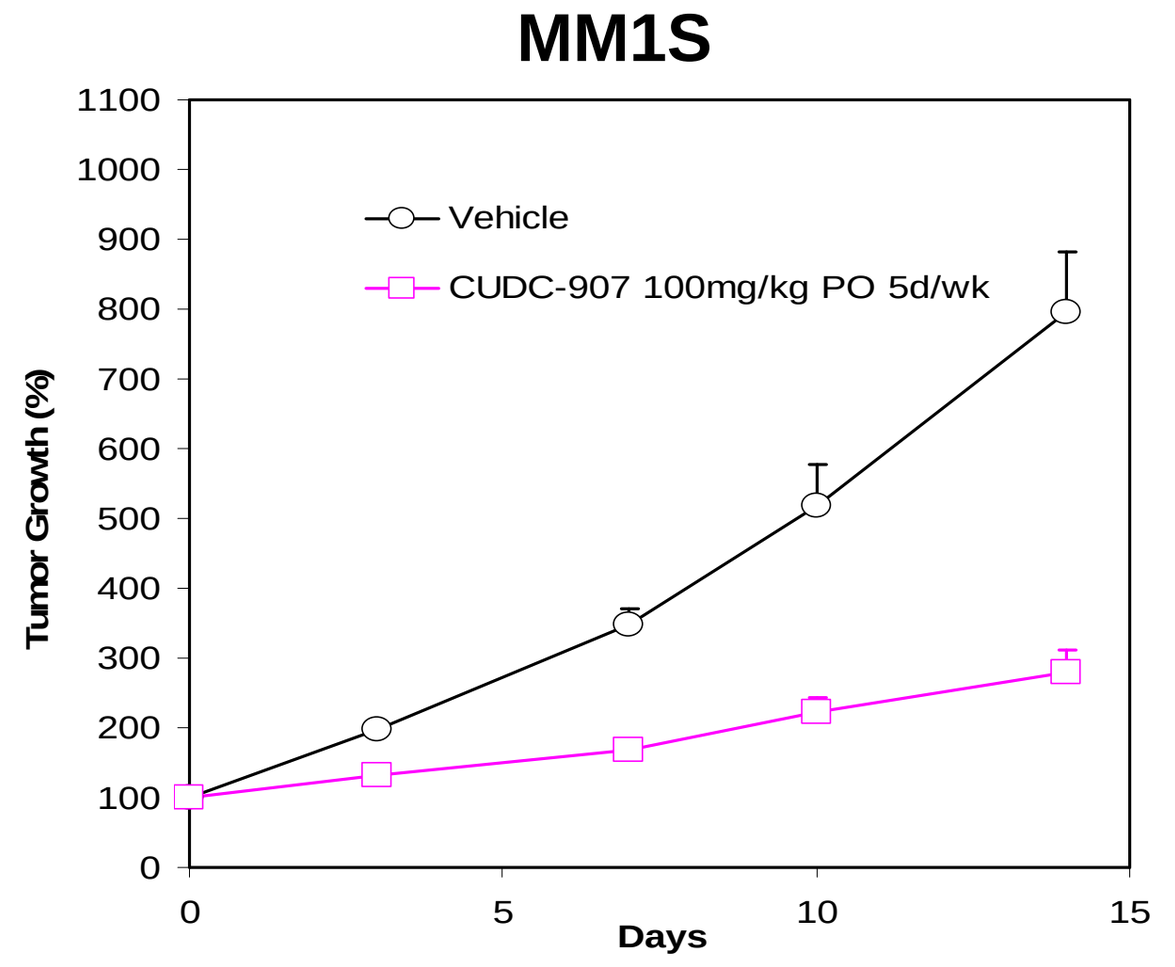
Vehicle
CUDC-907 100mg/kg PO 5d/wk
Tumor Growth (%)
Days

MM1R

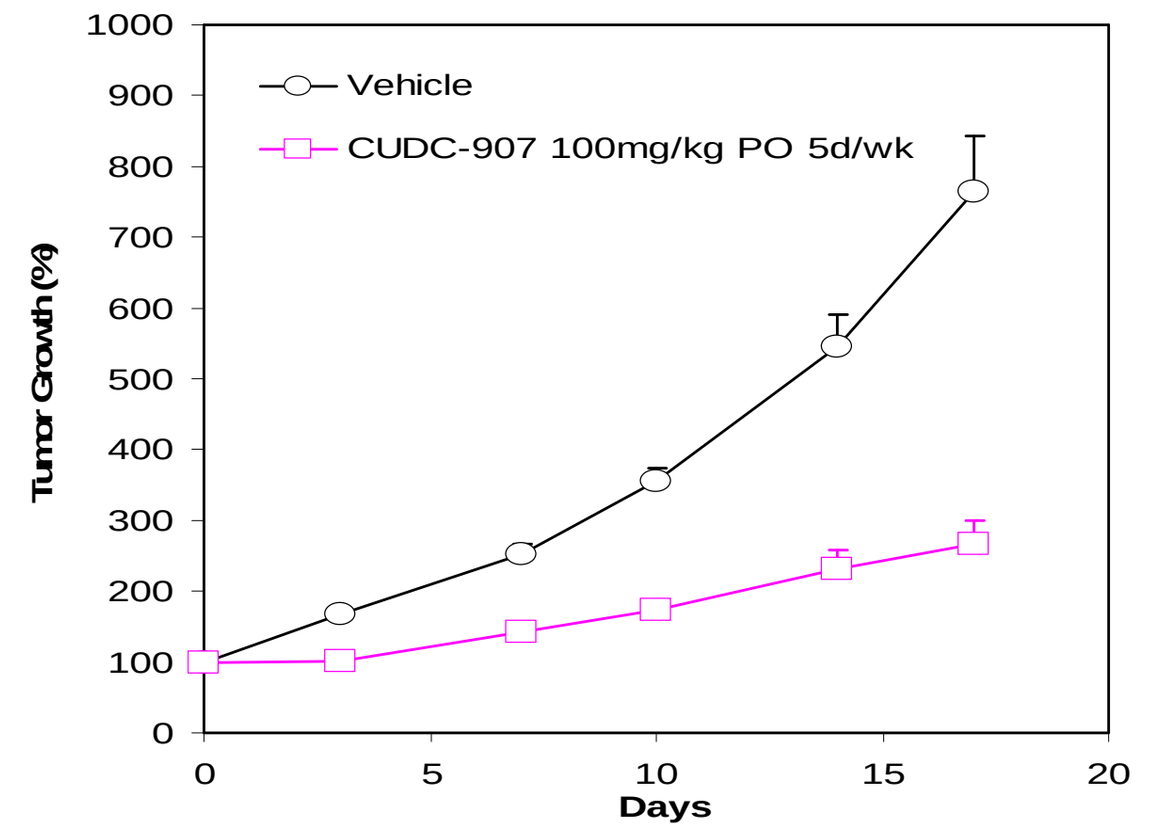
Vehicle
CUDC-907 100mg/kg PO 5d/wk
Days

MM1R

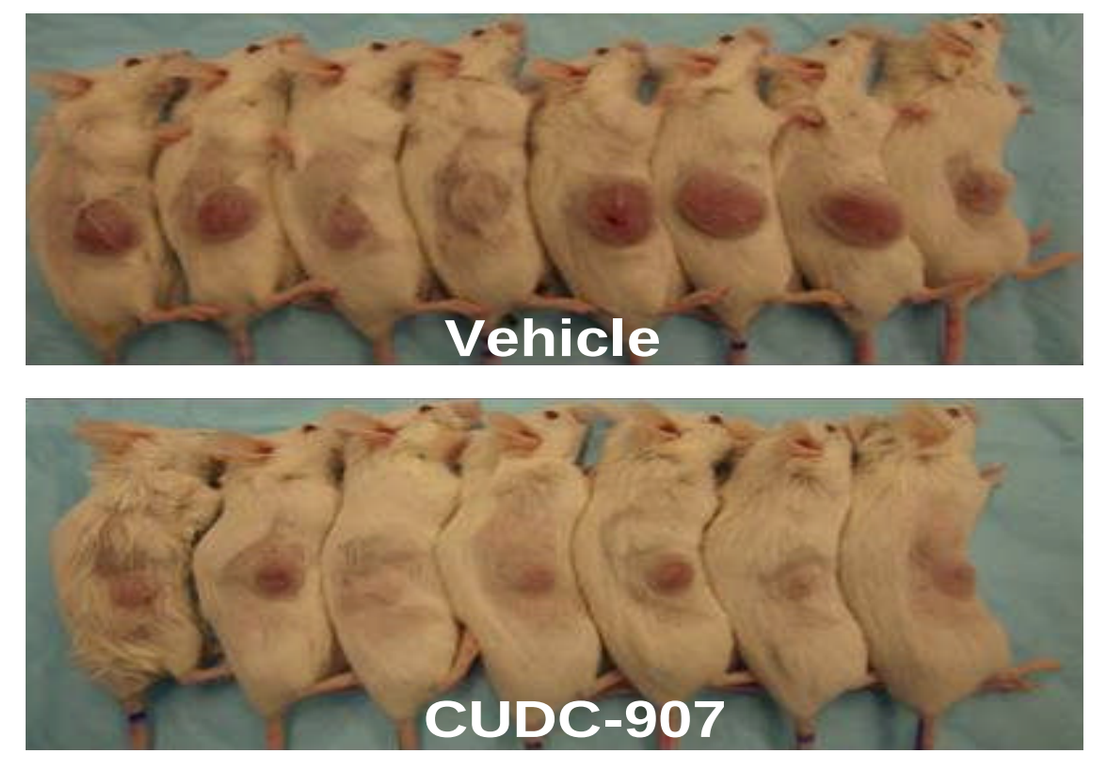
Vehicle
CUDC-907

CUDC-907 Efficacy in Various B Cell Lymphoma Models

SU-DHL4

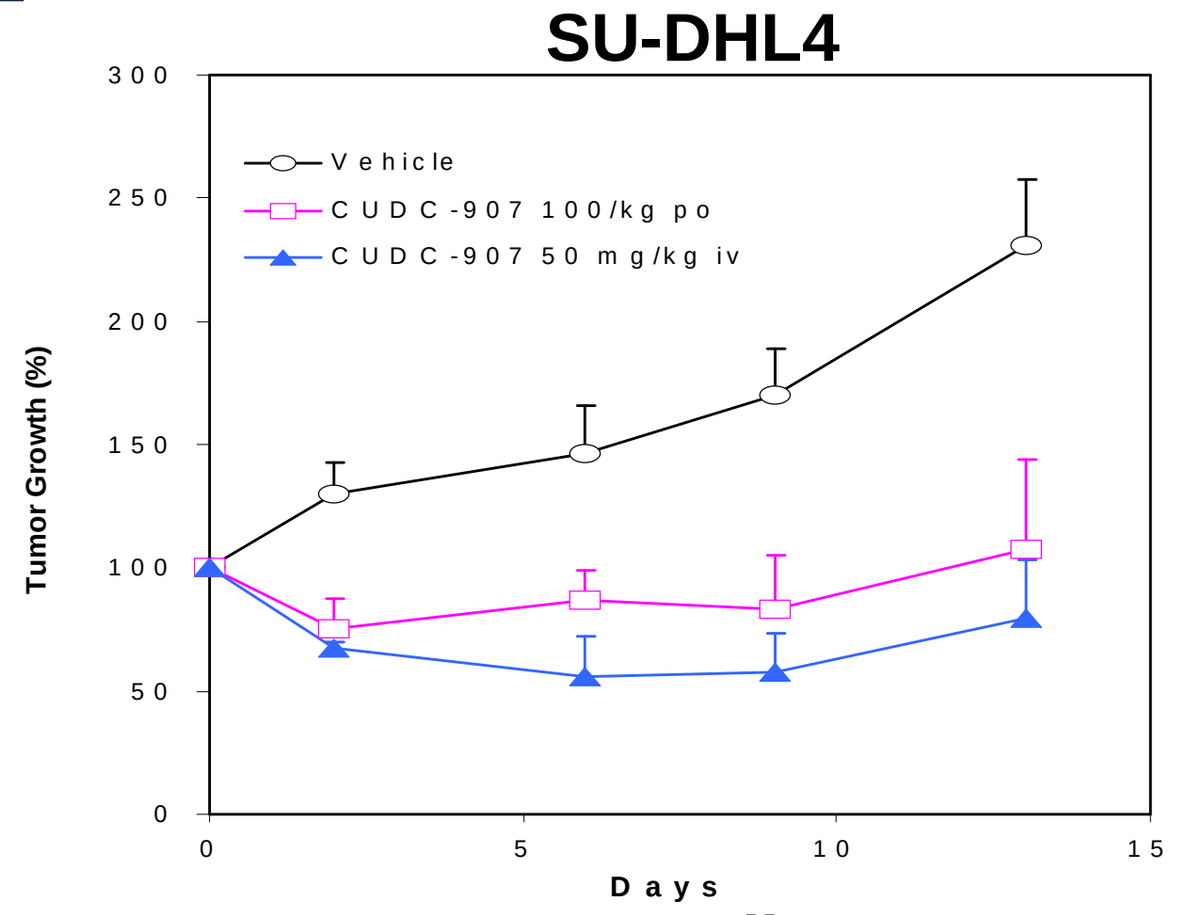
Vehicle
CUDC-907 100/kg po
CUDC-907 50 mg/kg iv
Tumor Growth (%)
Days

Daudi

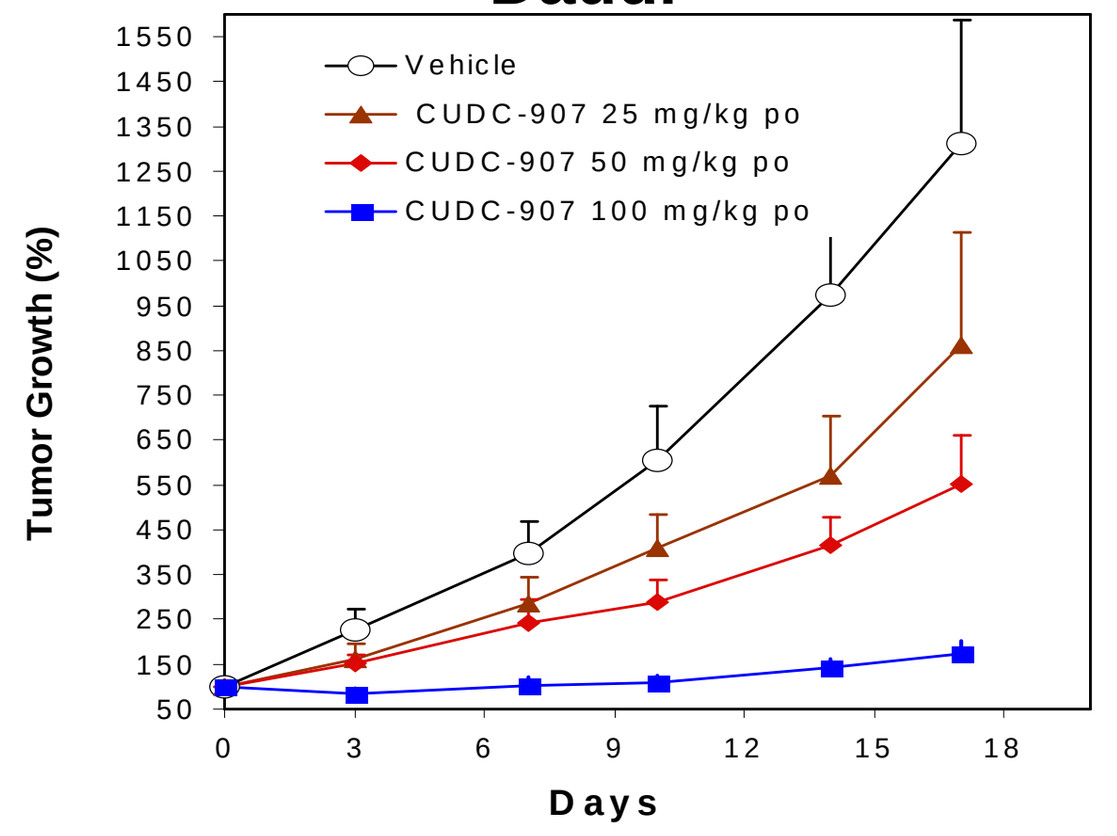
Vehicle
CUDC-907 25 mg/kg po
CUDC-907 50 mg/kg po
CUDC-907 100 mg/kg po
Tumor Growth (%)
Days

Daudi

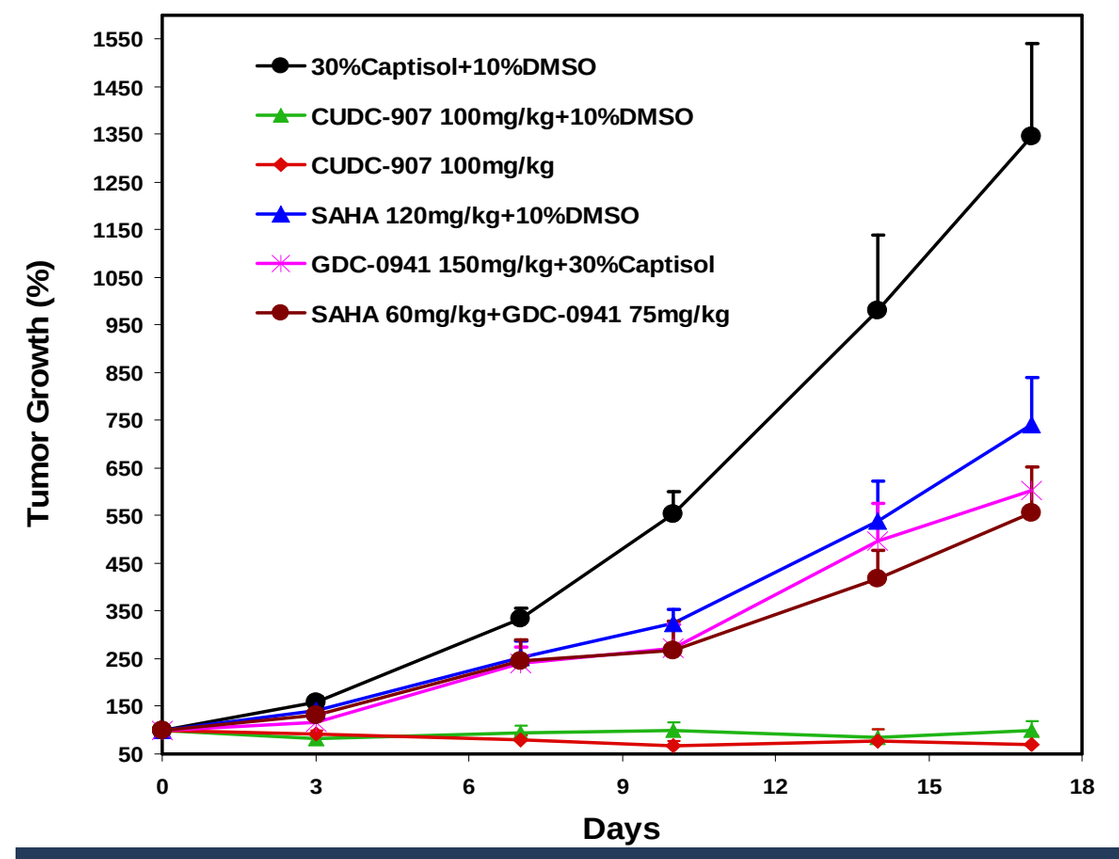
30%Captisol+10%DMSO
CUDC-907 100mg/kg+10%DMSO
CUDC-907 100mg/kg
SAHA 120mg/kg+10%DMSO
GDC-0941 150mg/kg+30%Captisol
SAHA 60mg/kg+GDC-0941 75mg/kg
Tumor Growth (%)
Days

Daudi

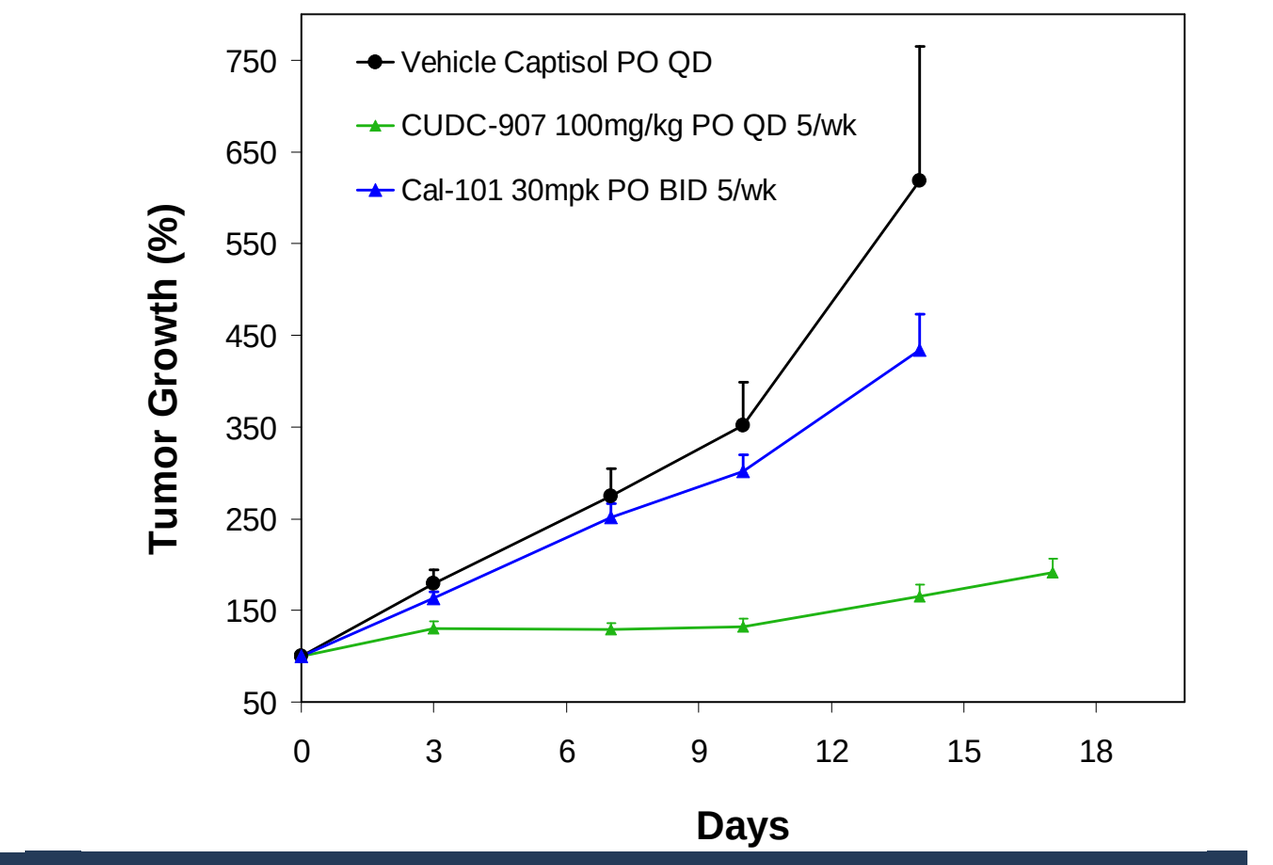
Vehicle Captisol PO QD
CUDC-907 100mg/kg PO QD 5/wk
Cal-101 30mpk PO BID 5/wk
Tumor Growth (%)
Days

Suboptimal Dose of CUDC-907 Combination Studies with SOC Agents in Various Models

MM1S

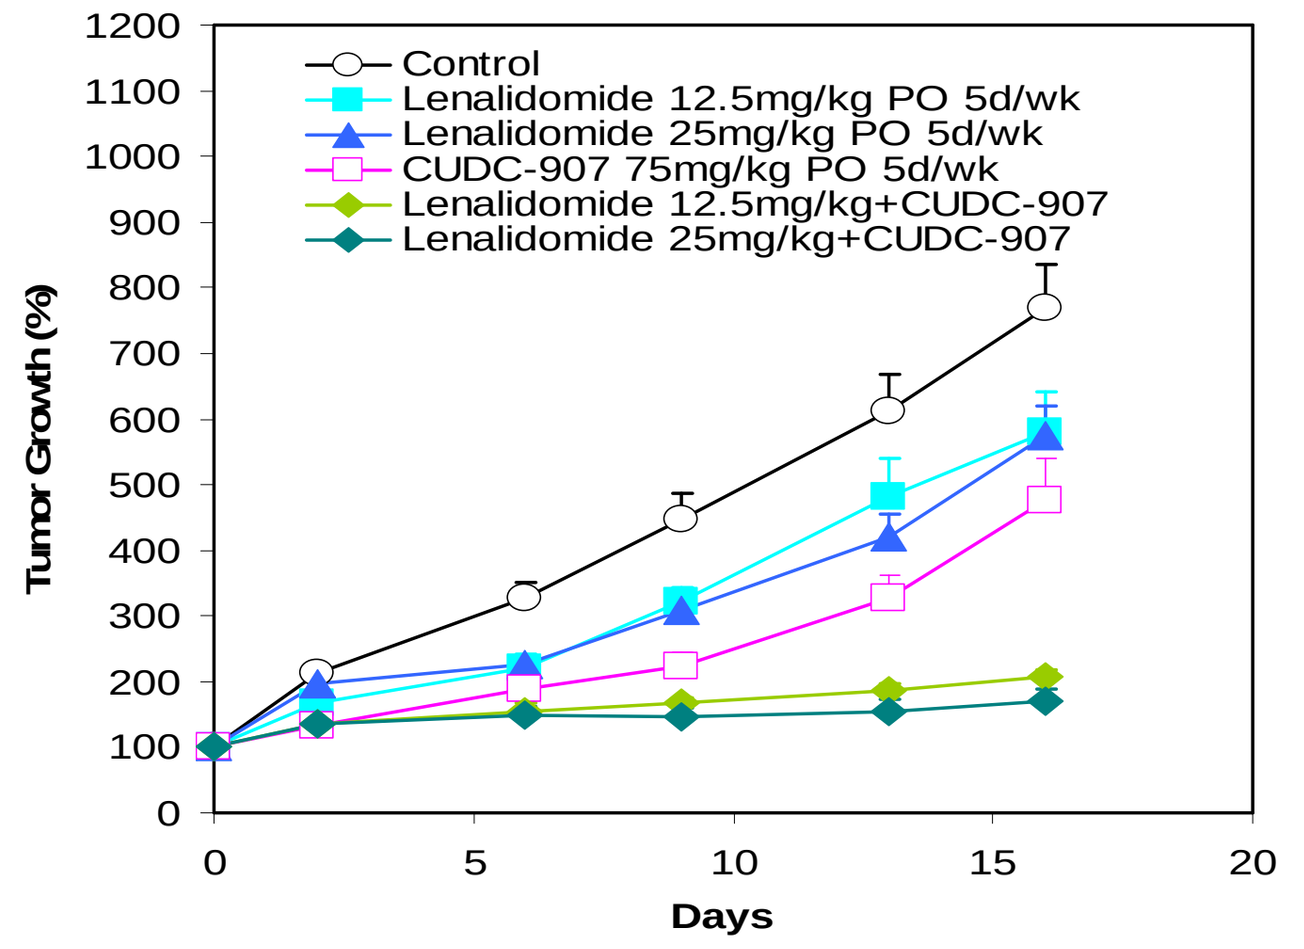
Control
Lenalidomide 12.5mg/kg PO 5d/wk
Lenalidomide 25mg/kg PO 5d/wk
CUDC-907 75mg/kg PO 5d/wk
Lenalidomide 12.5mg/kg+CUDC-907
Lenalidomide 25mg/kg+CUDC-907
Tumor Growth (%)
Days

Daudi

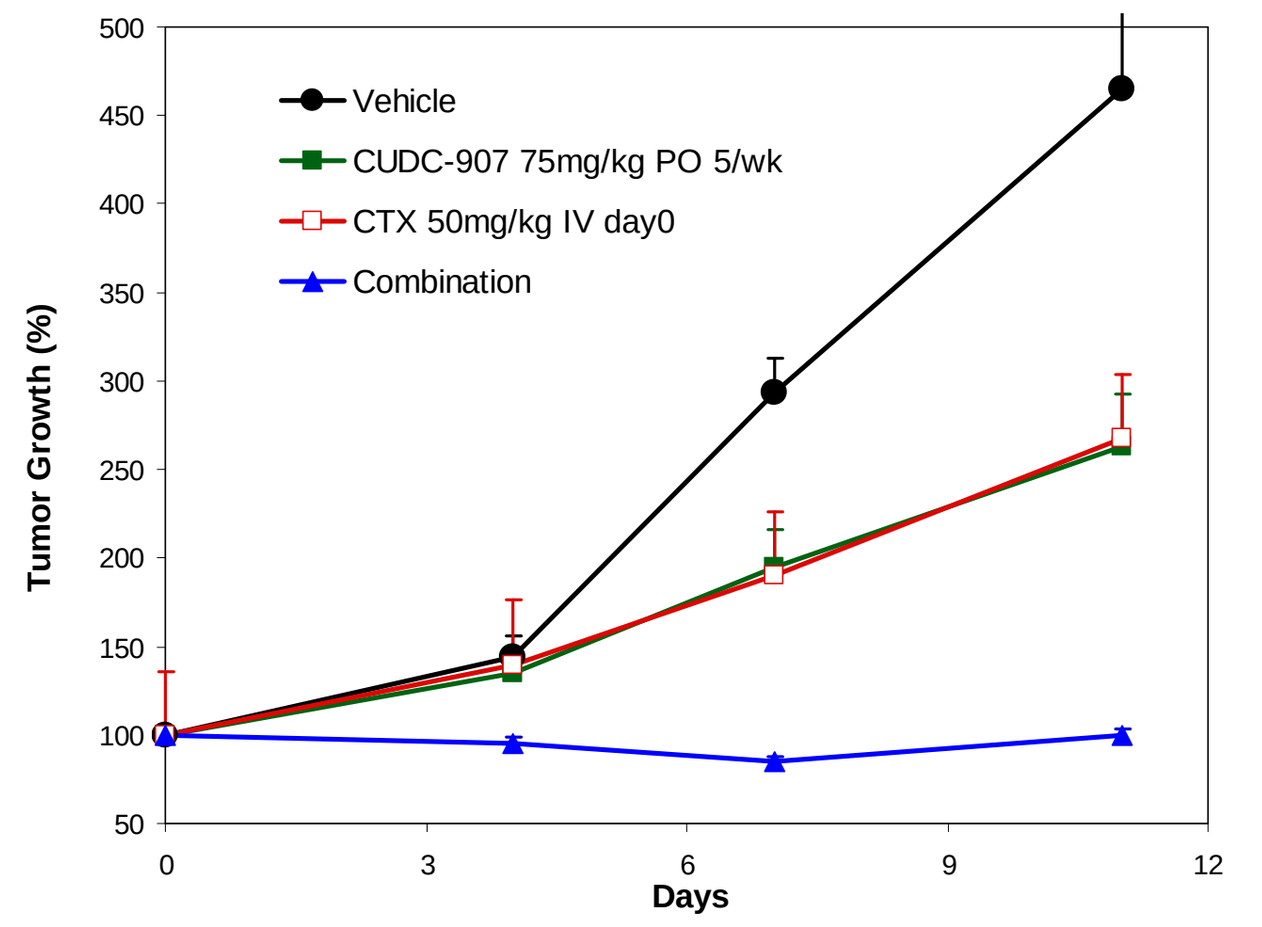
Vehicle
CUDC-907 75mg/kg PO 5/wk
CTX 50mg/kg IV day0
Combination
Tumor Growth (%)
Days

Expression of PI3K Isoforms in Human Hematological Cancer Xenografts

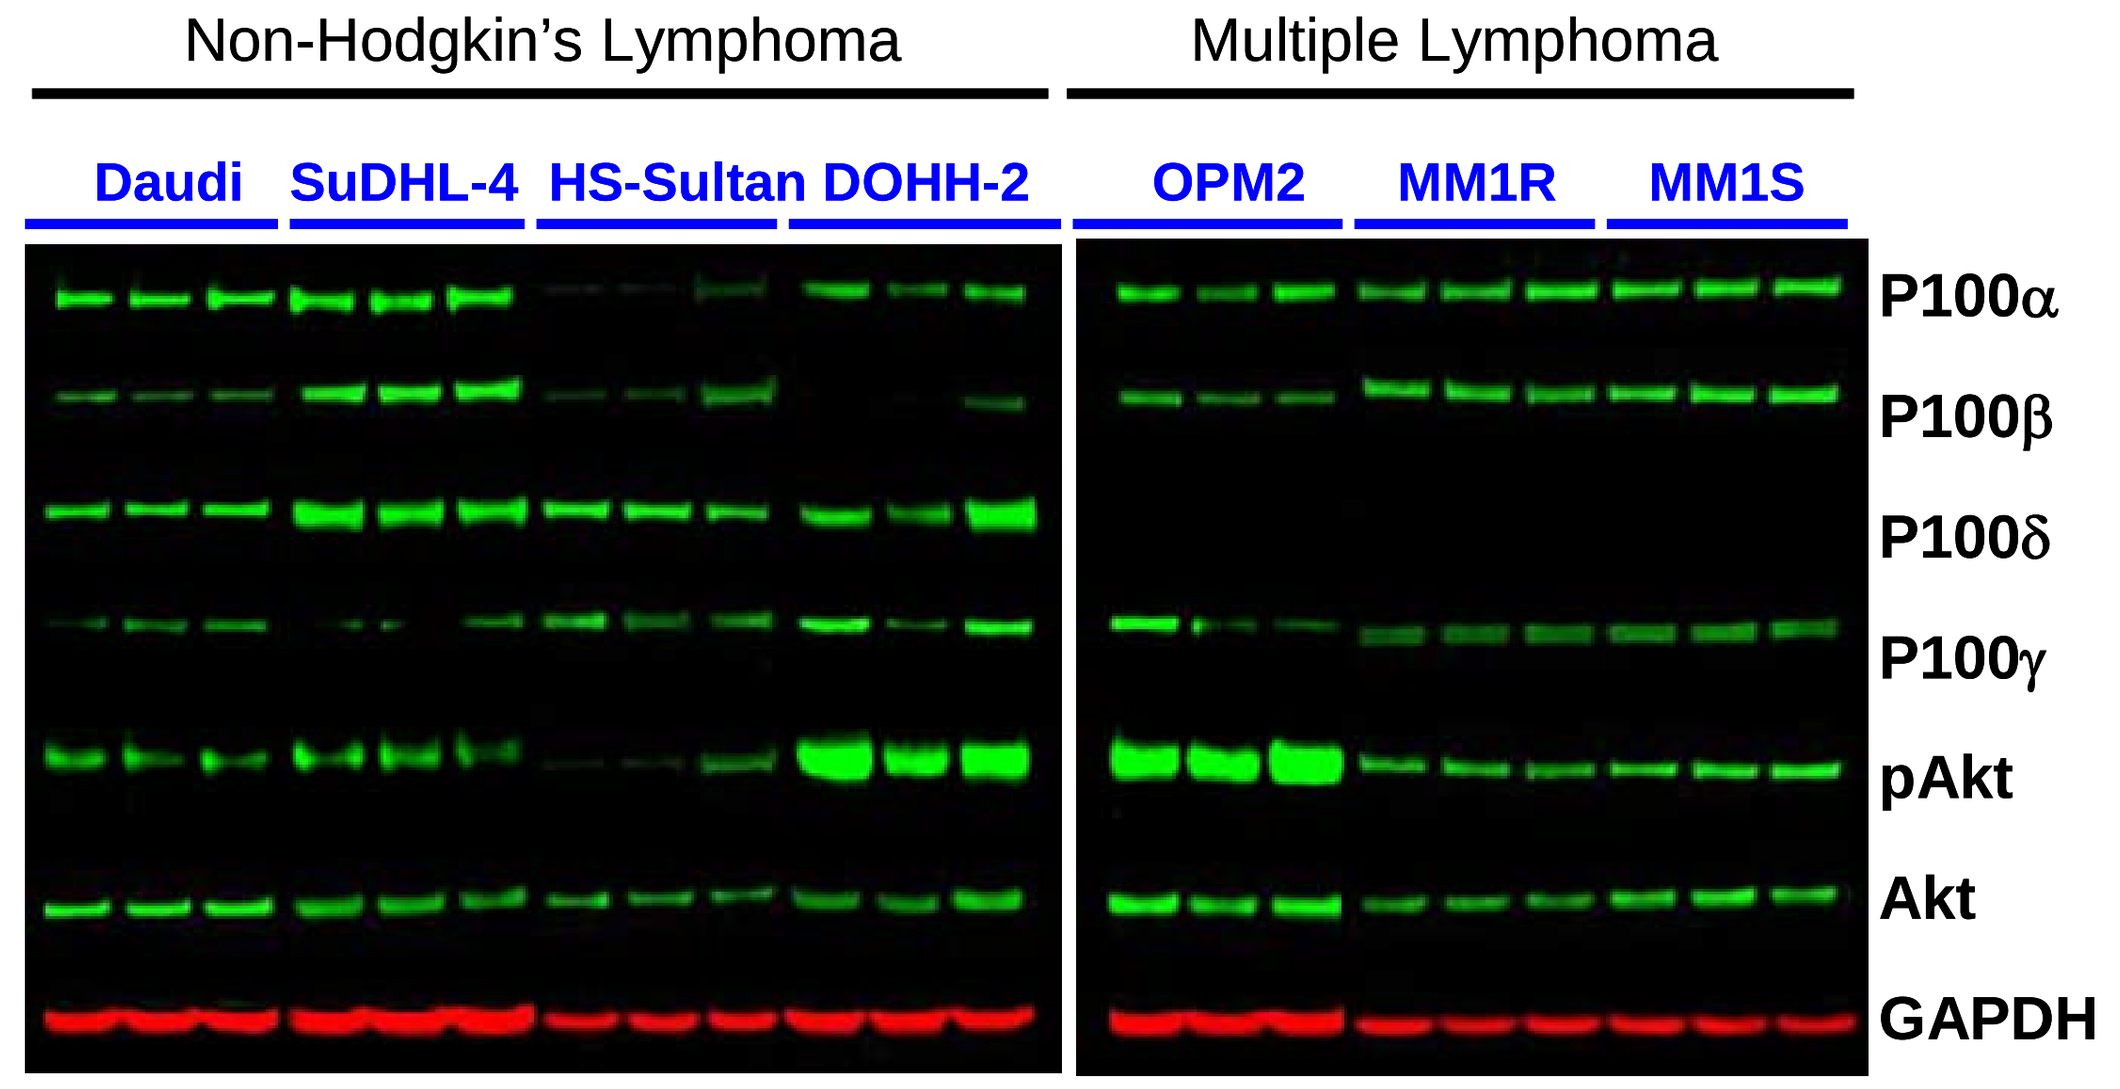
Non-Hodgkin's Lymphoma
Multiple Lymphoma
Daudi
SuDHL-4
HS-Sultan
DOHH-2
OPM2
MM1R
MM1S
P100α
P100β
P100δ
P100γ
pAkt
Akt
GAPDH

- Activation of AKT is frequently driven by more than one PI3K isoform
- Targeting more than one isoform may be more effective than the isoform selective PI3K inhibitors for hematological cancer

Conclusions

- CUDC-907 is a dual inhibitor of pan-HDAC and PI3K class-I
- CUDC-907 displays greater anti-tumor activity in multiple myeloma and B cell lymphoma in vitro and in vivo than reference compounds
- CUDC-907 is able to enhance antitumor activity when combined with standard of care agents in multiple myeloma and B cell lymphoma xenograft models
- In contrast to other isoform-selective PI3K inhibitors, CUDC-907 targets more than one PI3K isoform which might be required for a more effective treatment of hematological cancers
- CUDC-907 was selected as a candidate for clinical development.