Efficacy of Novel IRAK4 Inhibitors in ABC-DLBCL Xenograft Models

Wesley Roy Balasubramanian, Venkateshwar Rao Gummadi, Kavitha Nellore, Sivapriya Marappan, Aravind Basavaraju, Bharathi Raja Ainan, Girish Daginakatte, Sreevalsam Gopinath, Sanjeev Giri, Thomas Antony, Shekar Chelur, Susanta Samajdar, Chetan Pandit, Murali Ramachandra
Aurigene Discovery Technologies Ltd, Bangalore, India

Abstract

Interleukin-1 receptor associated kinases (IRAKs) are serine/threonine protein kinases belonging to tyrosine-like kinase (TLK) family. The IRAK family consists of IRAK1, IRAK2, IRAK3 and IRAK4 out of which only IRAK1 and IRAK4 exhibit kinase activity. IRAKs function as mediators of Toll-like receptor (TLR) and interleukin-1 receptor (IL-1R) signaling pathways and play an important role in innate immune signaling. Recent studies have reported the occurrence of oncogenic mutations in MYD88 in ~30% of activated B cell diffuse large B-cell lymphoma (ABC DLBCL) and ~90% of Waldenstrom's macroglobulinemia (WM) leading to constitutive activation of the IRAK4 and NFkB pathway. Thus IRAKs are attractive therapeutic targets for treatment of malignancies with altered innate immune signaling such as ABC DLBCL. We have designed, synthesized and tested small molecule inhibitors of IRAK4 based on hits originating from Aurigene's compound library. We have identified a series of novel bicyclic heterocycles as potent inhibitors of IRAK4 with moderate to very high selectivity (S35 score = 0.03) in a 329 kinase panel. Lead compounds were profiled in proliferation and mechanistic assays (p-IRAK1 inhibition) in appropriate ABC DLBCL cell lines. Aurigene lead compounds demonstrate potent inhibition of cellular proliferation with a good correlation to inhibition of phosphorylation of signaling intermediates in mechanistic assays. Lead compounds exhibit excellent PK profile and good oral bioavailability in rodents. Lead compounds demonstrated dose dependent inhibition of TLR4 induced TNFα release in-vivo. Selected compounds demonstrate excellent in-vivo efficacy in a ABC DLBCL xenograft model with >90% tumor growth inhibition and good in-vivo PD modulation. Preliminary in-vitro toxicology studies as well as observations in MTD study indicate a clean safety profile. In summary, a series of potent and selective IRAK4 inhibitors have been discovered and are being evaluated for treatment of cancers with dysregulated innate immune signaling.

IRAK Inhibitors for treatment of ABC DLBCL

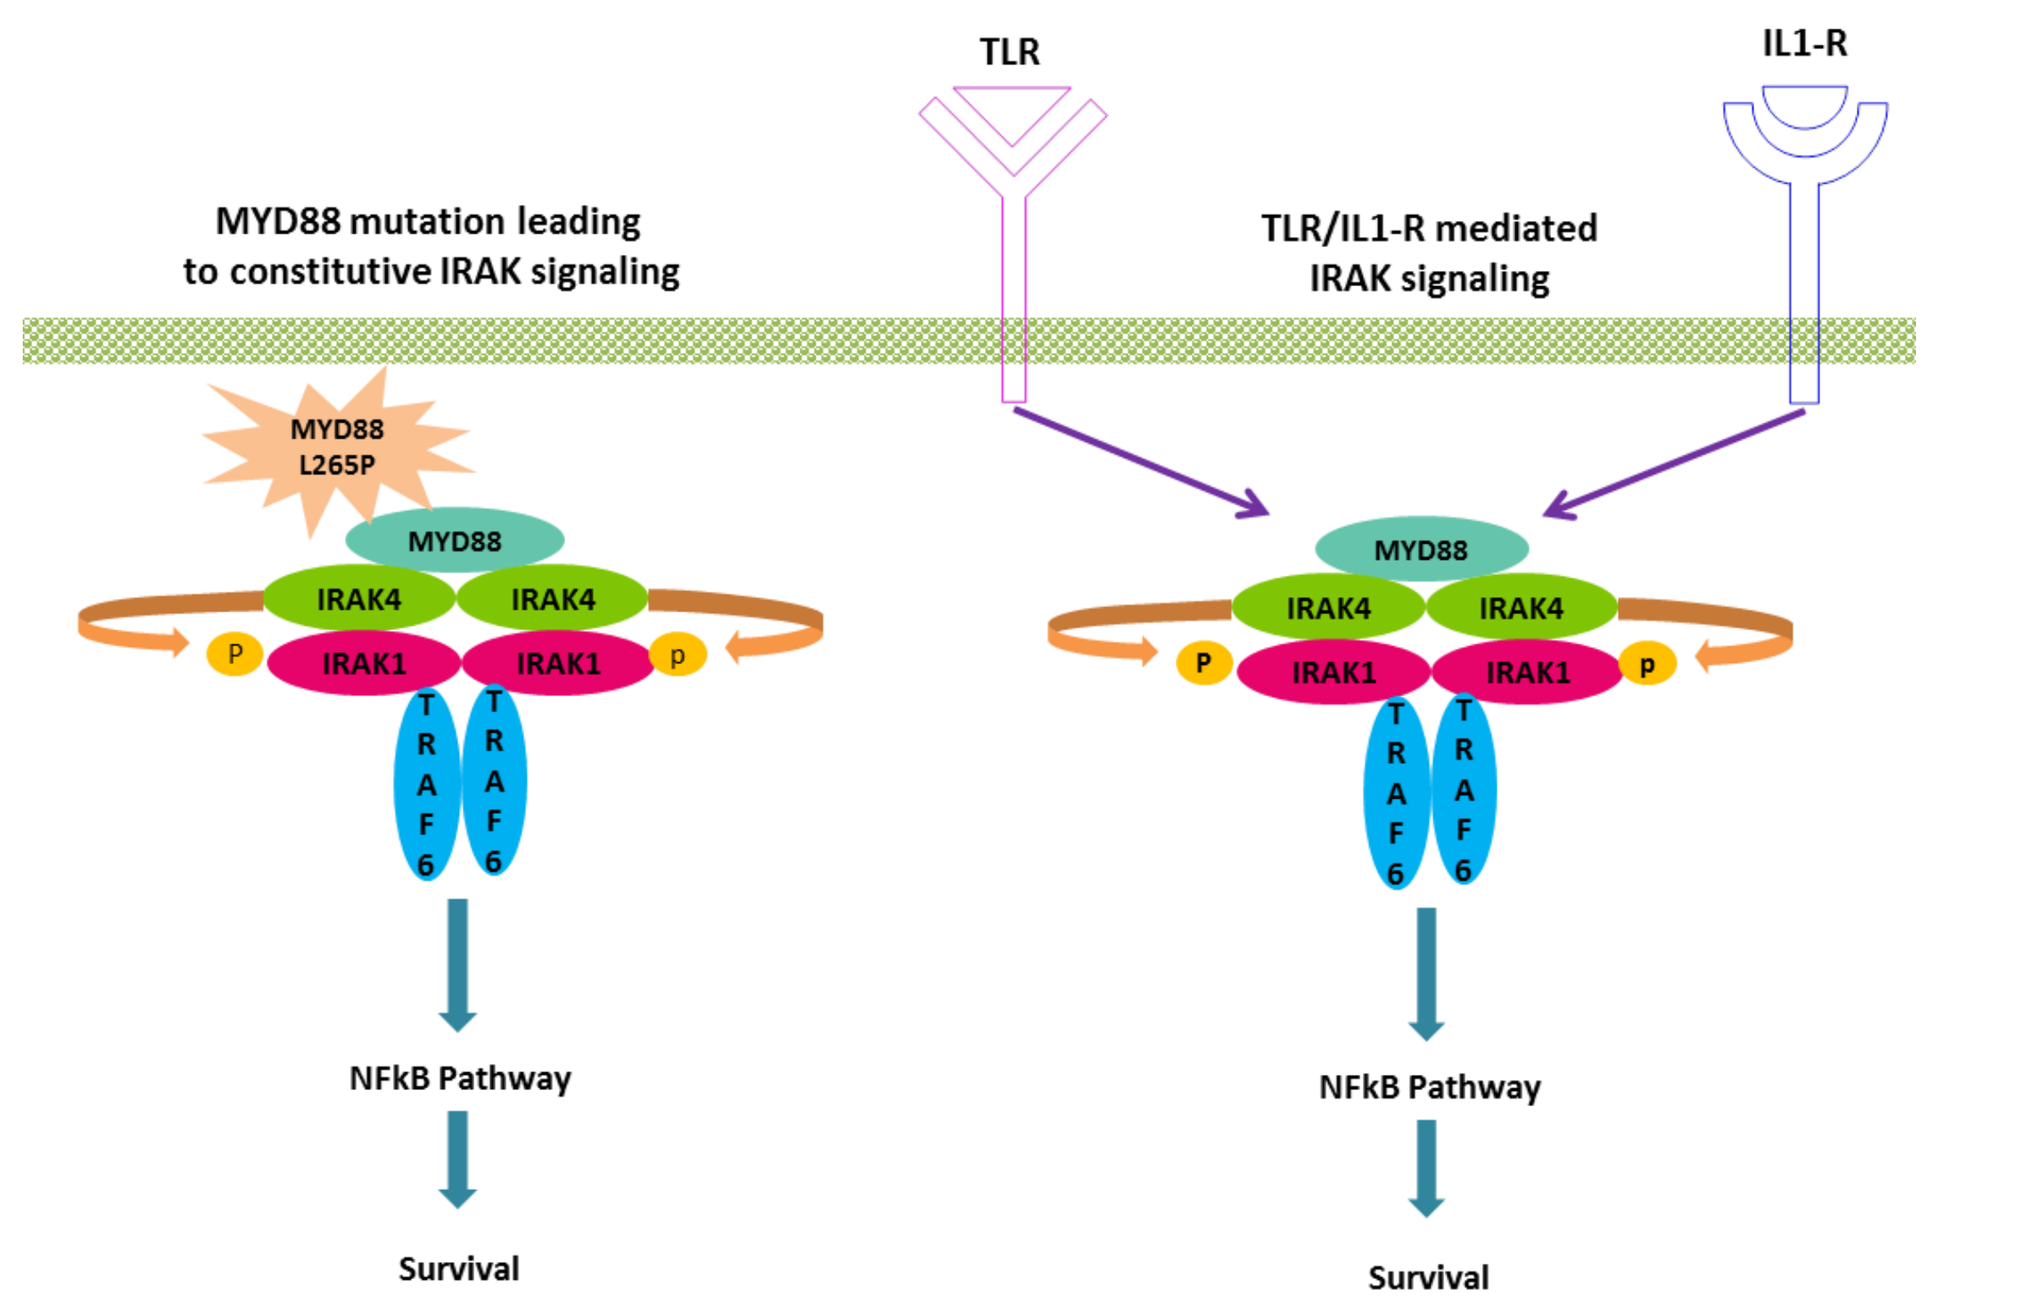
TLR
IL1-R
MYD88 mutation leading to constitutive IRAK signaling
TLR/IL1-R mediated IRAK signaling
MYD88 L265P
MYD88
IRAK4
IRAK1
P
TRAF6
NFkB Pathway
Survival

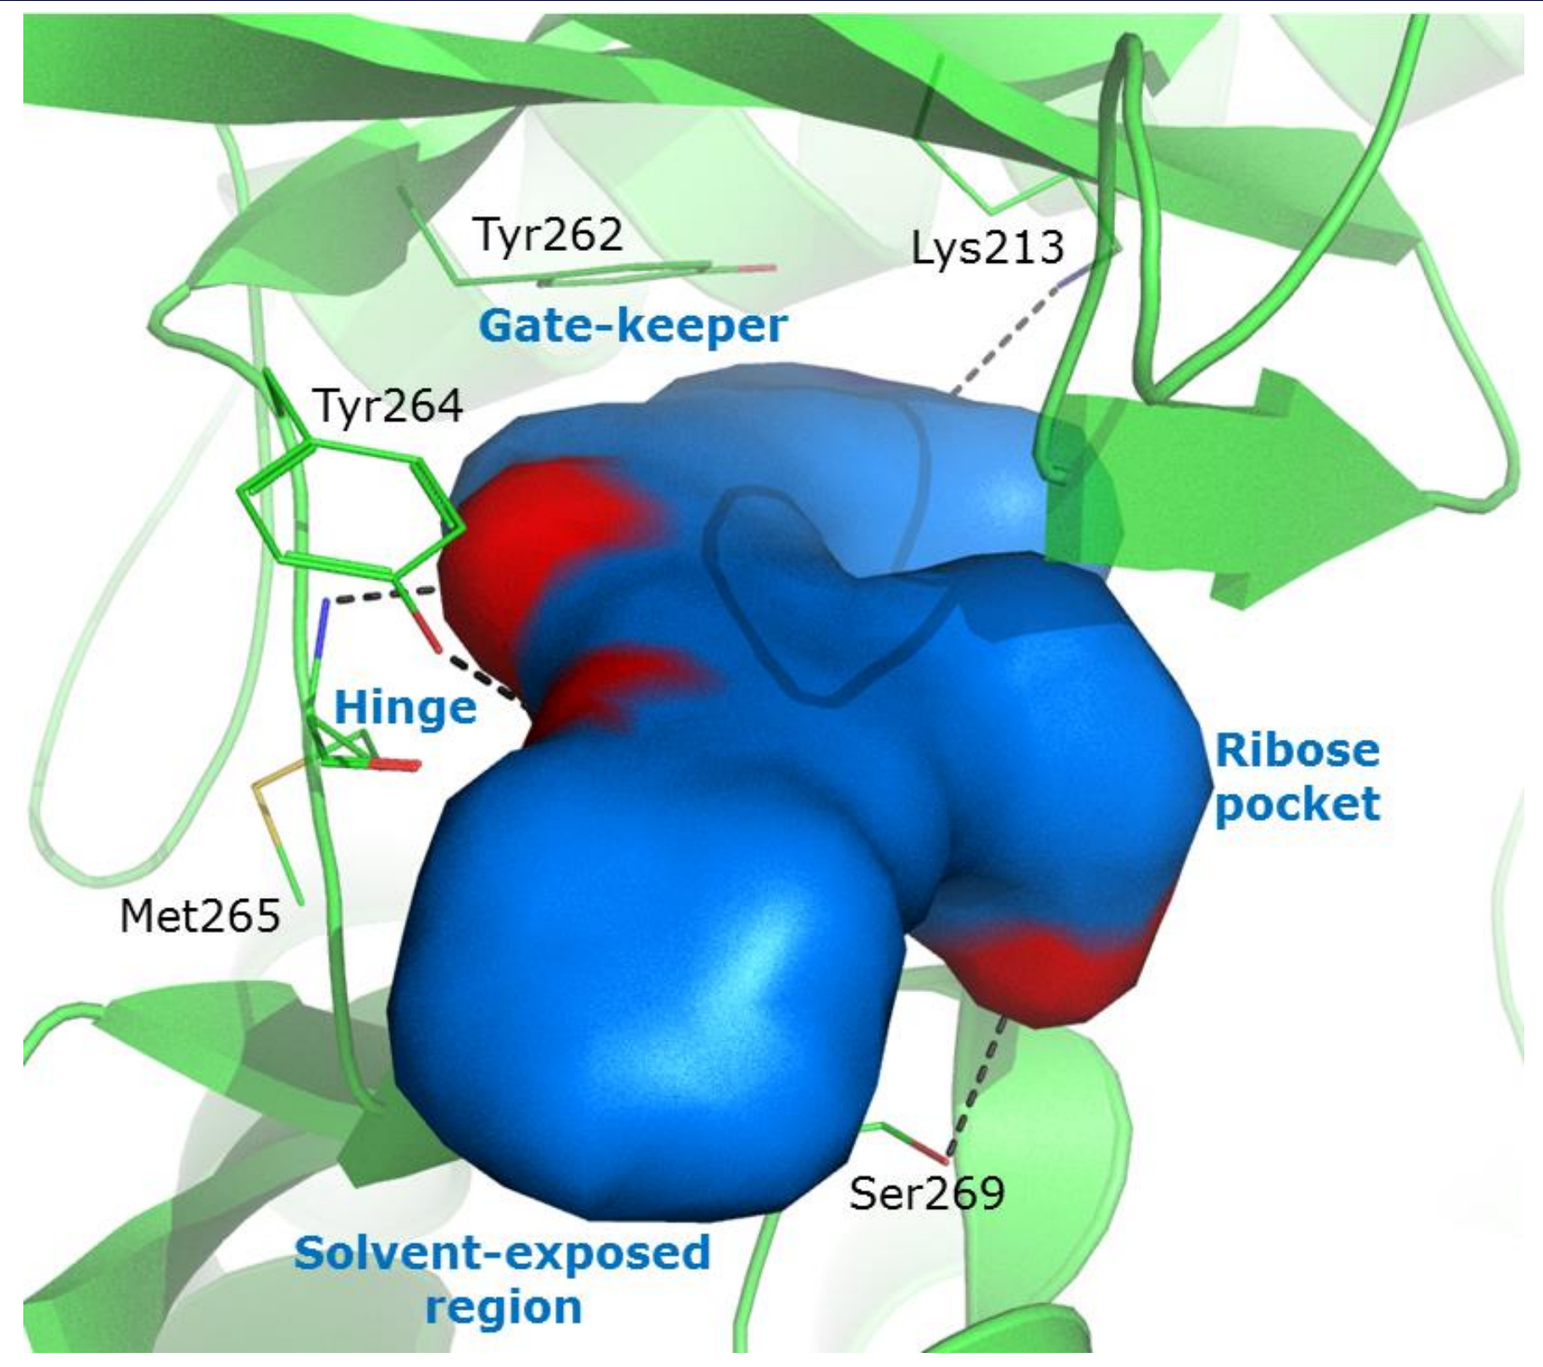
Tyr262
Gate-keeper
Lys213
Tyr264
Hinge
Met265
Ribose pocket
Ser269
Solvent-exposed region

- MYD88 adaptor protein transduces receptor signaling to IRAK4
- ~30% ABC-DLBCL, ~90% WM and ~6% CLL patients have activating MYD88 mutations
- Activating MYD88 mutations lead to constitutive activation of IRAK signaling
- Aurigene's lead compound docked with hIRAK4-kinase domain
- Compound anticipated to bind in ATP binding pocket close to gate-keeper residue (Tyr262)
- Polar interactions with hinge residues (Tyr264 & Met265) and back-pocket catalytic lysine (Lys213)

Potency and Selectivity Profile

Compound	hIRAK4 IC_{50} nM	Kinase Selectivity S35 Score	
		at 1µM	at 10µM
AU-4948	37	0.0343	0.0515
AU-6686	3.5	0.518	0.615

AU-4948 exhibits best-in-class selectivity, comparable to the most selective kinase inhibitors in market

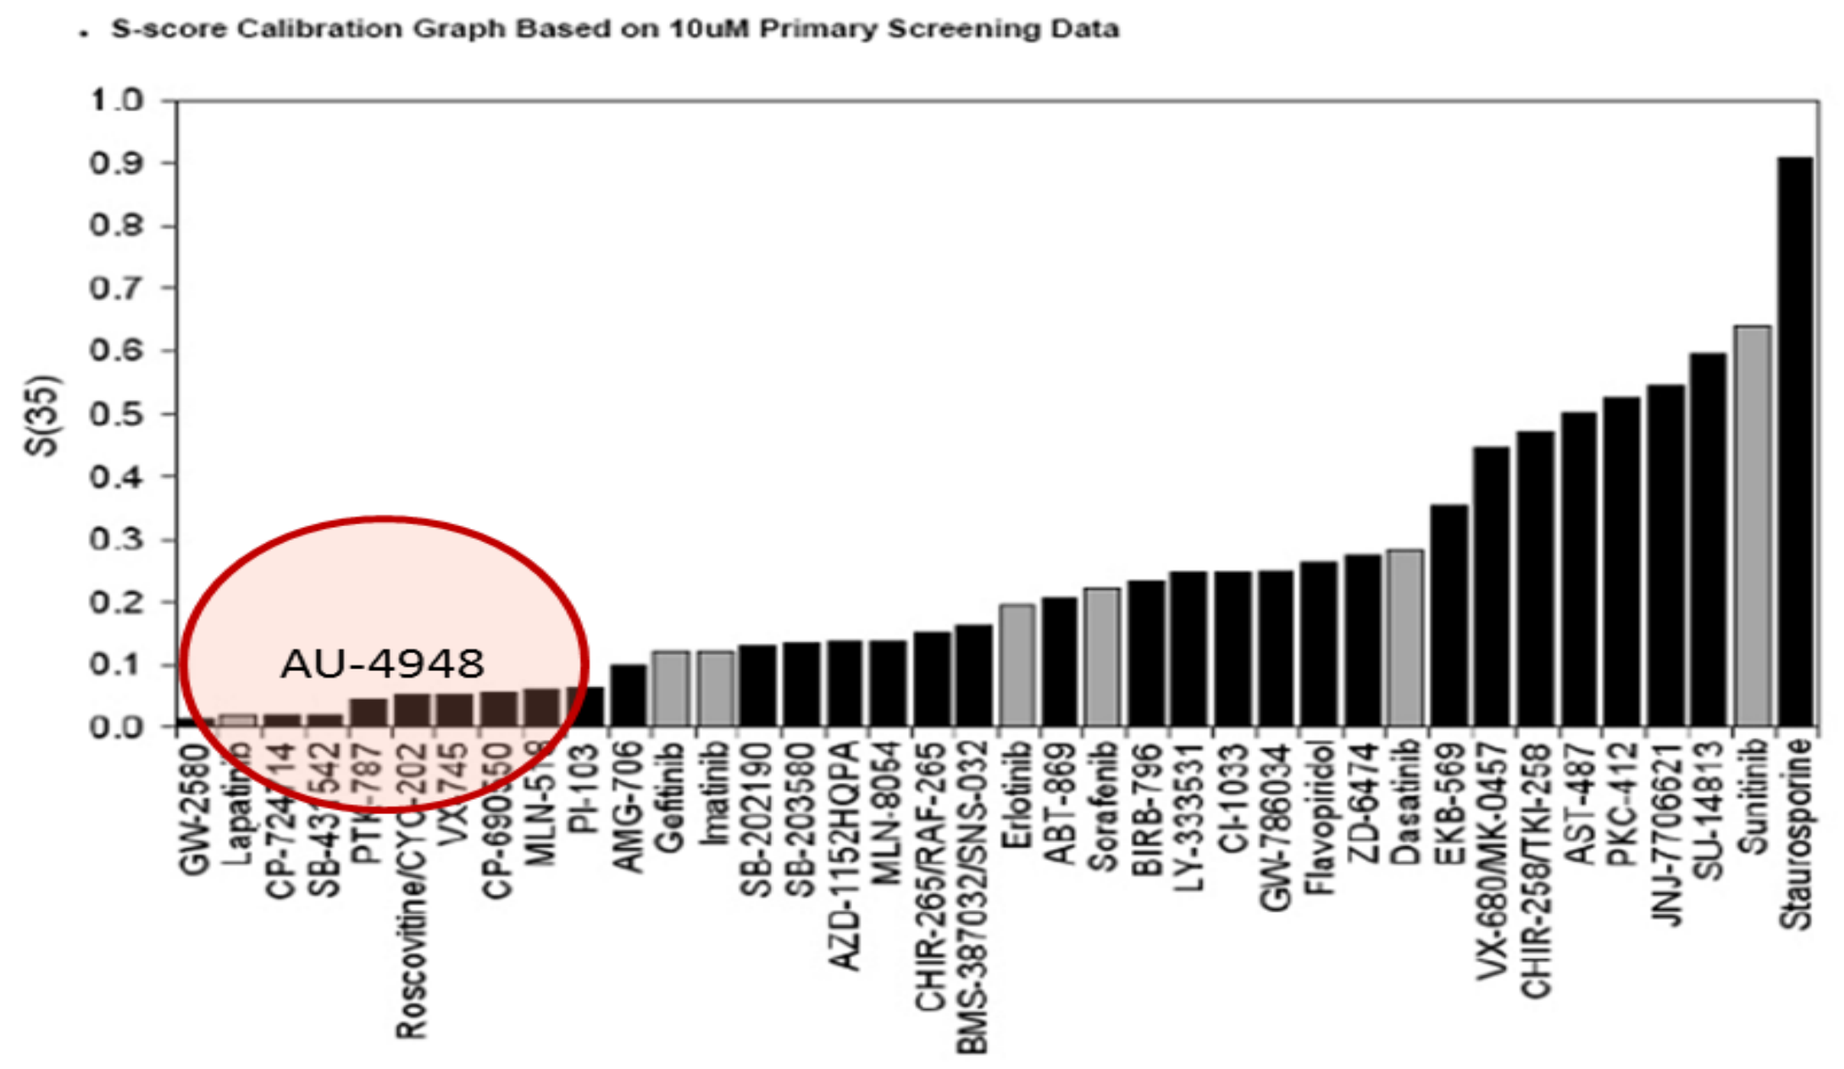
S-score Calibration Graph Based on 10uM Primary Screening Data
S(35)
AU-4948
GW-2580
Lapatinib
CP-724714
SB-431542
PTK-787
Roscovitine/CYC-202
VX-745
CP-690550
MLN-518
PI-103
AMG-706
Gefitinib
Imatinib
SB-202190
SB-203580
AZD-1152HQPA
MLN-8054
CHIR-265/RAF-265
BMS-387032/SNS-032
Erlotinib
ABT-869
Sorafenib
BIRB-796
LY-333531
CI-1033
GW-786034
Flavopiridol
ZD-6474
Dasatinib
EKB-569
VX-680/MK-0457
CHIR-258/TKI-258
AST-487
PKC-412
JNJ-7706621
SU-14813
Sunitinib
Staurosporine

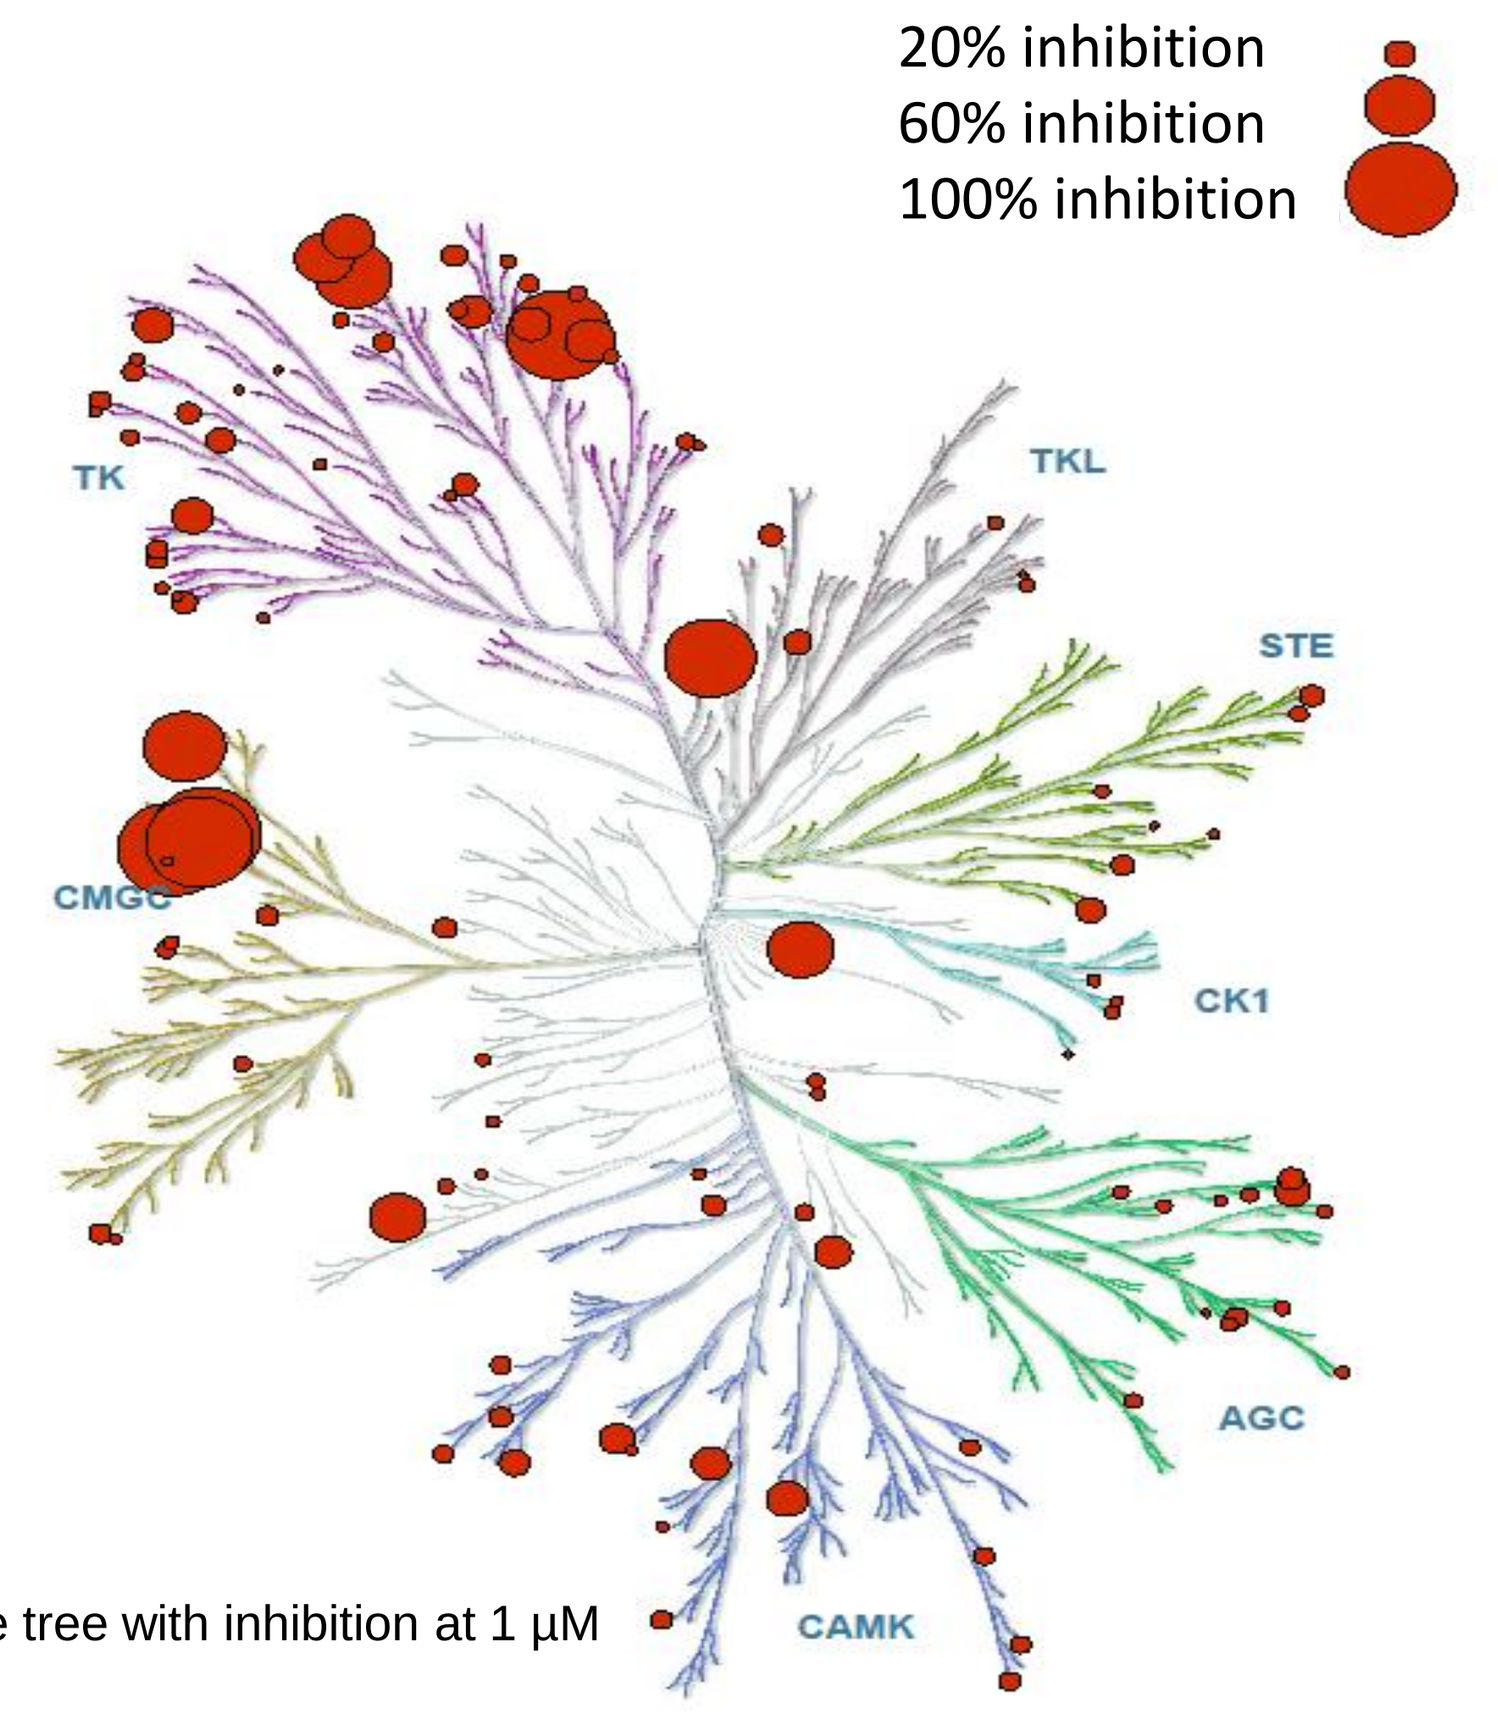
20% inhibition
60% inhibition
100% inhibition
TK
TKL
STE
CMGC
CK1
AGC
CAMK

Kinome tree with inhibition at 1 µM

"Illustration reproduced courtesy of Cell Signaling Technology, Inc. (www.cellsignal.com)"

DMPK Profile of Lead Compounds

Parameters		AU-6686	AU-4948
Thermodynamic Solubility, pH7.4 (µM)		<2	39
Stability in MLM, HLM, RLM $t_{1/2}$ (min); Clin (µl/min/mg)		>60;<38(all species)	>60;<38(all species)
CaCO2 Permeability		Low	Medium
PPB (%bound) Rat /Mouse /Human		>99.9 (all species)	99.7/ 92.7/77.5
Mouse IV PK Dose: 3 mg/kg	$t_{1/2}$ (hr)	5.97	2.58
	$AUC_{(0-inf)}$ (ng.hr/mL)	2516	5393
	Cl(mL/min/kg)	22.53	9.27
	Vd_{ss} (L/kg)	12.94	0.66
Mouse Oral PK Dose: 10 mg/kg	C_{max} (ng/mL)	1274	6618
	$AUC_{(0-inf)}$ (ng.hr/mL)	6170	12741
	F(%)	74	71

In-vivo activity of AU-4948 in TLR4 induced TNFα release

AU-4948, po — Theophylline i.p — LPS 0.3 mg/kg, i.p. — Blood collection

Wistar Rats: - 3 hr, -0.5 hr, 0 hr, 1hr

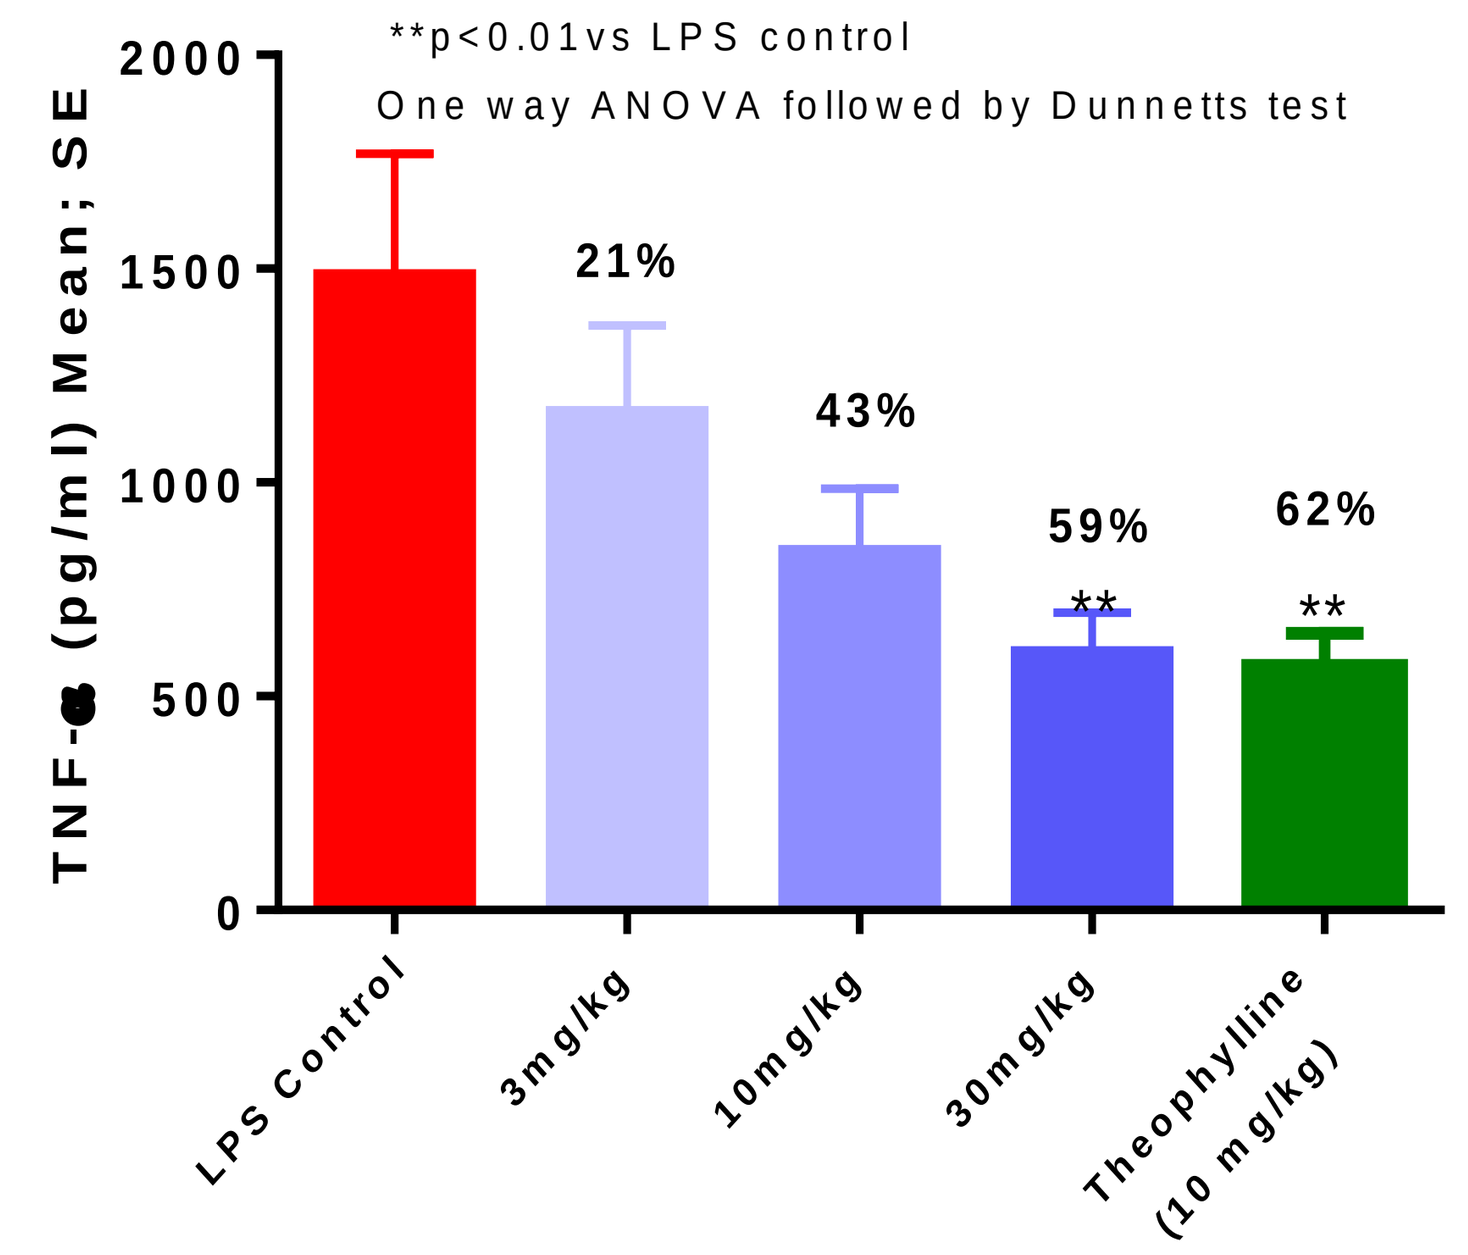
**p<0.01vs LPS control
One way ANOVA followed by Dunnetts test
TNF-α (pg/ml) Mean; SE
2000
1500
1000
500
0
21%
43%
59%
62%
**
**
LPS Control
3mg/kg
10mg/kg
30mg/kg
Theophylline (10 mg/kg)

Group / Dose of AU-4948	% Reduction	4 hr plasma compound levels (ng/mL)
LPS Control	--	--
3 mg/kg	21	2441.3 ± 492.4
10 mg/kg	43	6407.9 ± 1209.8
30 mg/kg	59	18167.9 ± 1379.4

- Dose dependent reduction of LPS induced TNF-α levels with AU-4948 treatment

Efficacy of AU-6686 and AU-4948 in OCI-Ly3 Xenograft Model

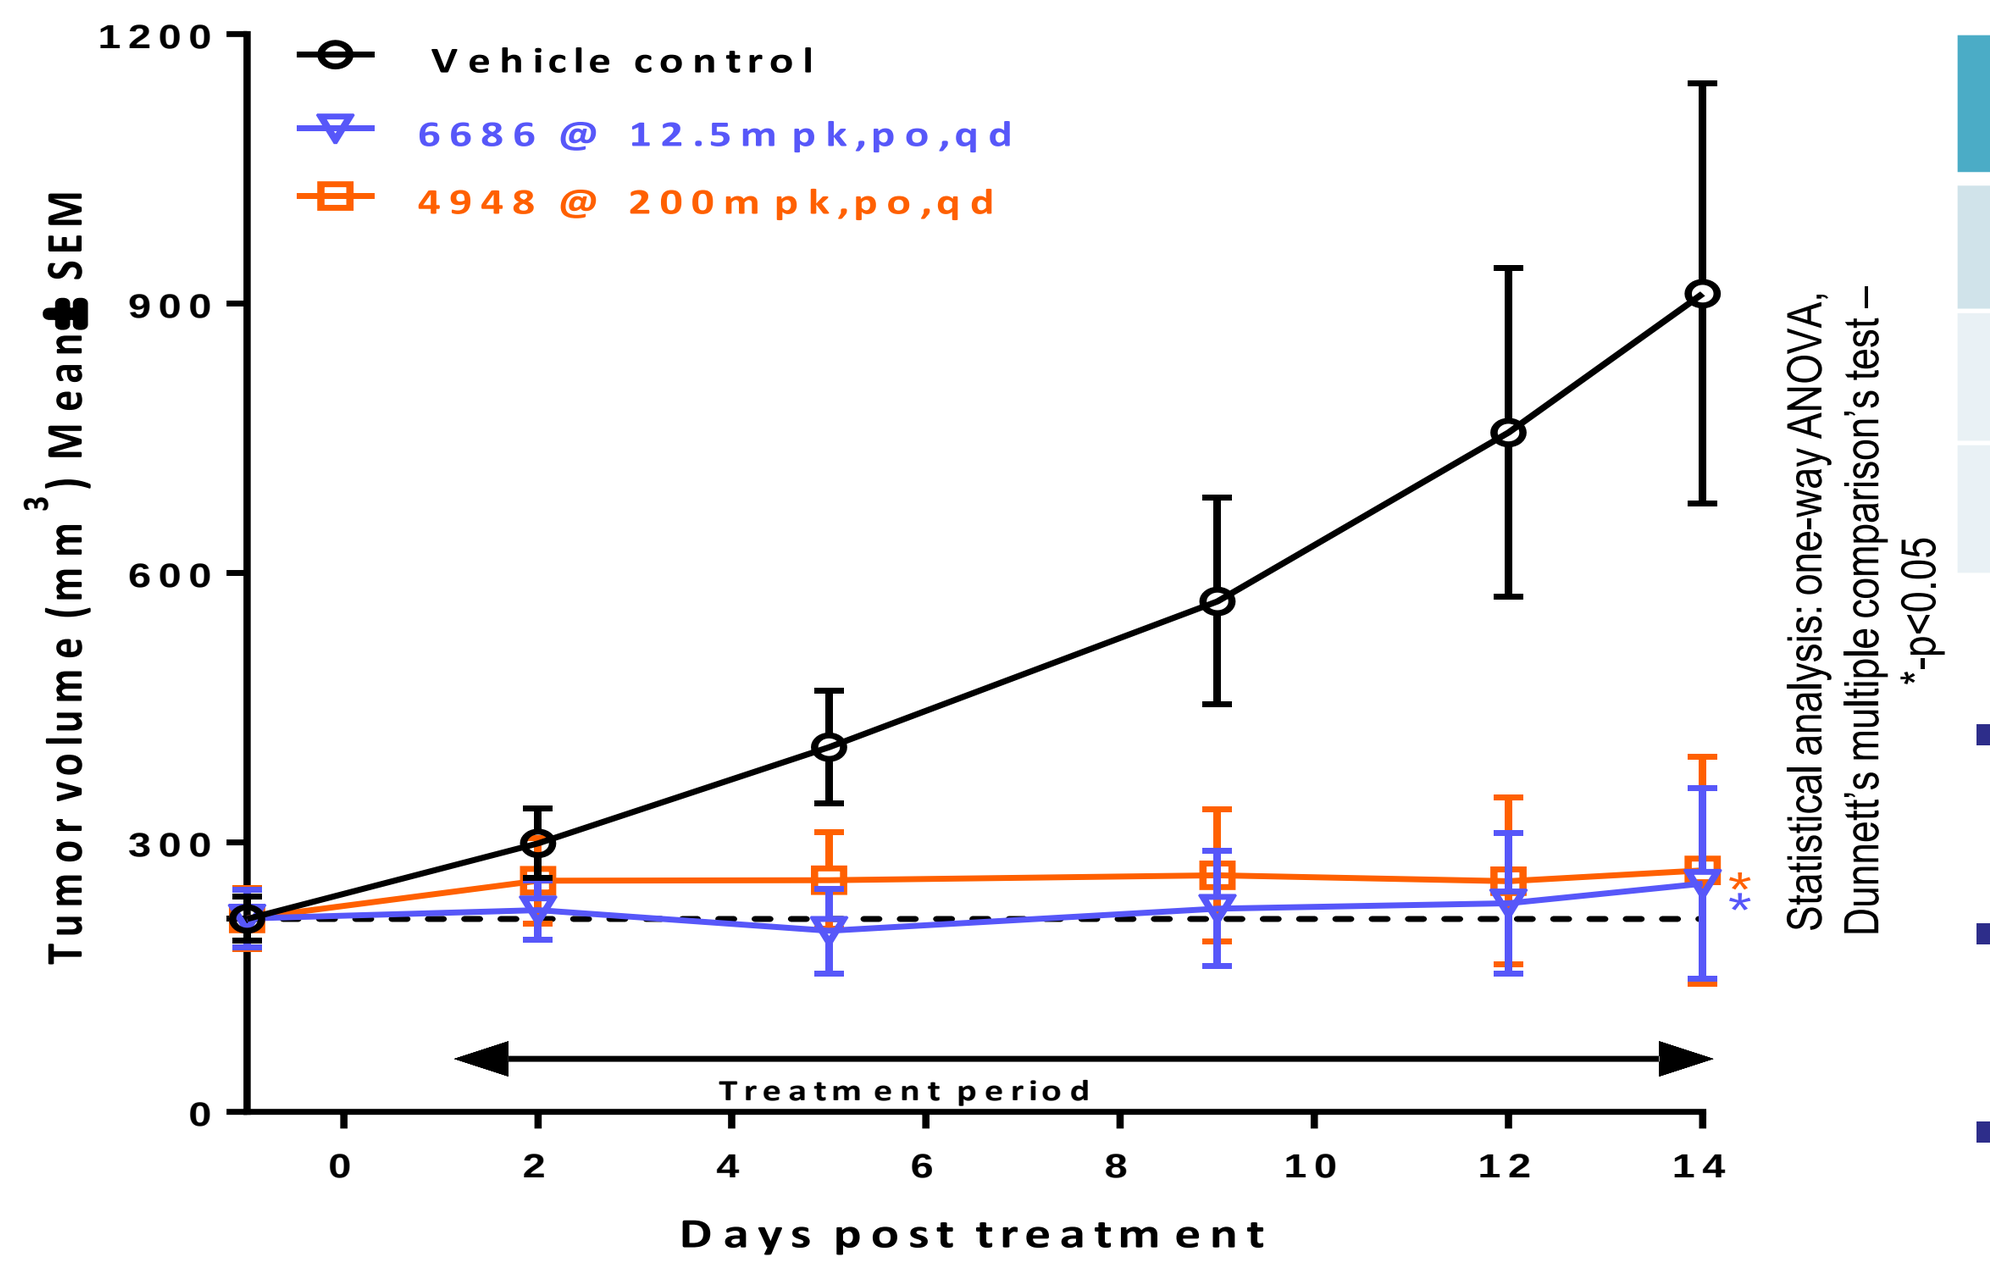
Vehicle control
6686 @ 12.5mpk,po,qd
4948 @ 200mpk,po,qd
Tumor volume (mm3) Mean ± SEM
1200
900
600
300
0
Treatment period
0
2
4
6
8
10
12
14
Days post treatment
Statistical analysis: one-way ANOVA, Dunnett's multiple comparison's test – *-p<0.05

Treatment groups (n=8)	%TGI (Day-14)
Vehicle control, po,qd	NA
AU-6686 @ 12.5 mpk, po,qd	94*
AU-4948 @ 200 mpk, po,qd	92*

- OCI-Ly3 is ABC-DLBCL with MYD88-L265P mutation
- AU-6686 at 12.5 mpk and AU-4948 at 200 mpk showed significant tumor growth inhibition
- No significant body weight loss in any treatment group

Efficacy of AU-4948 in OCI-Ly3 Xenograft Model

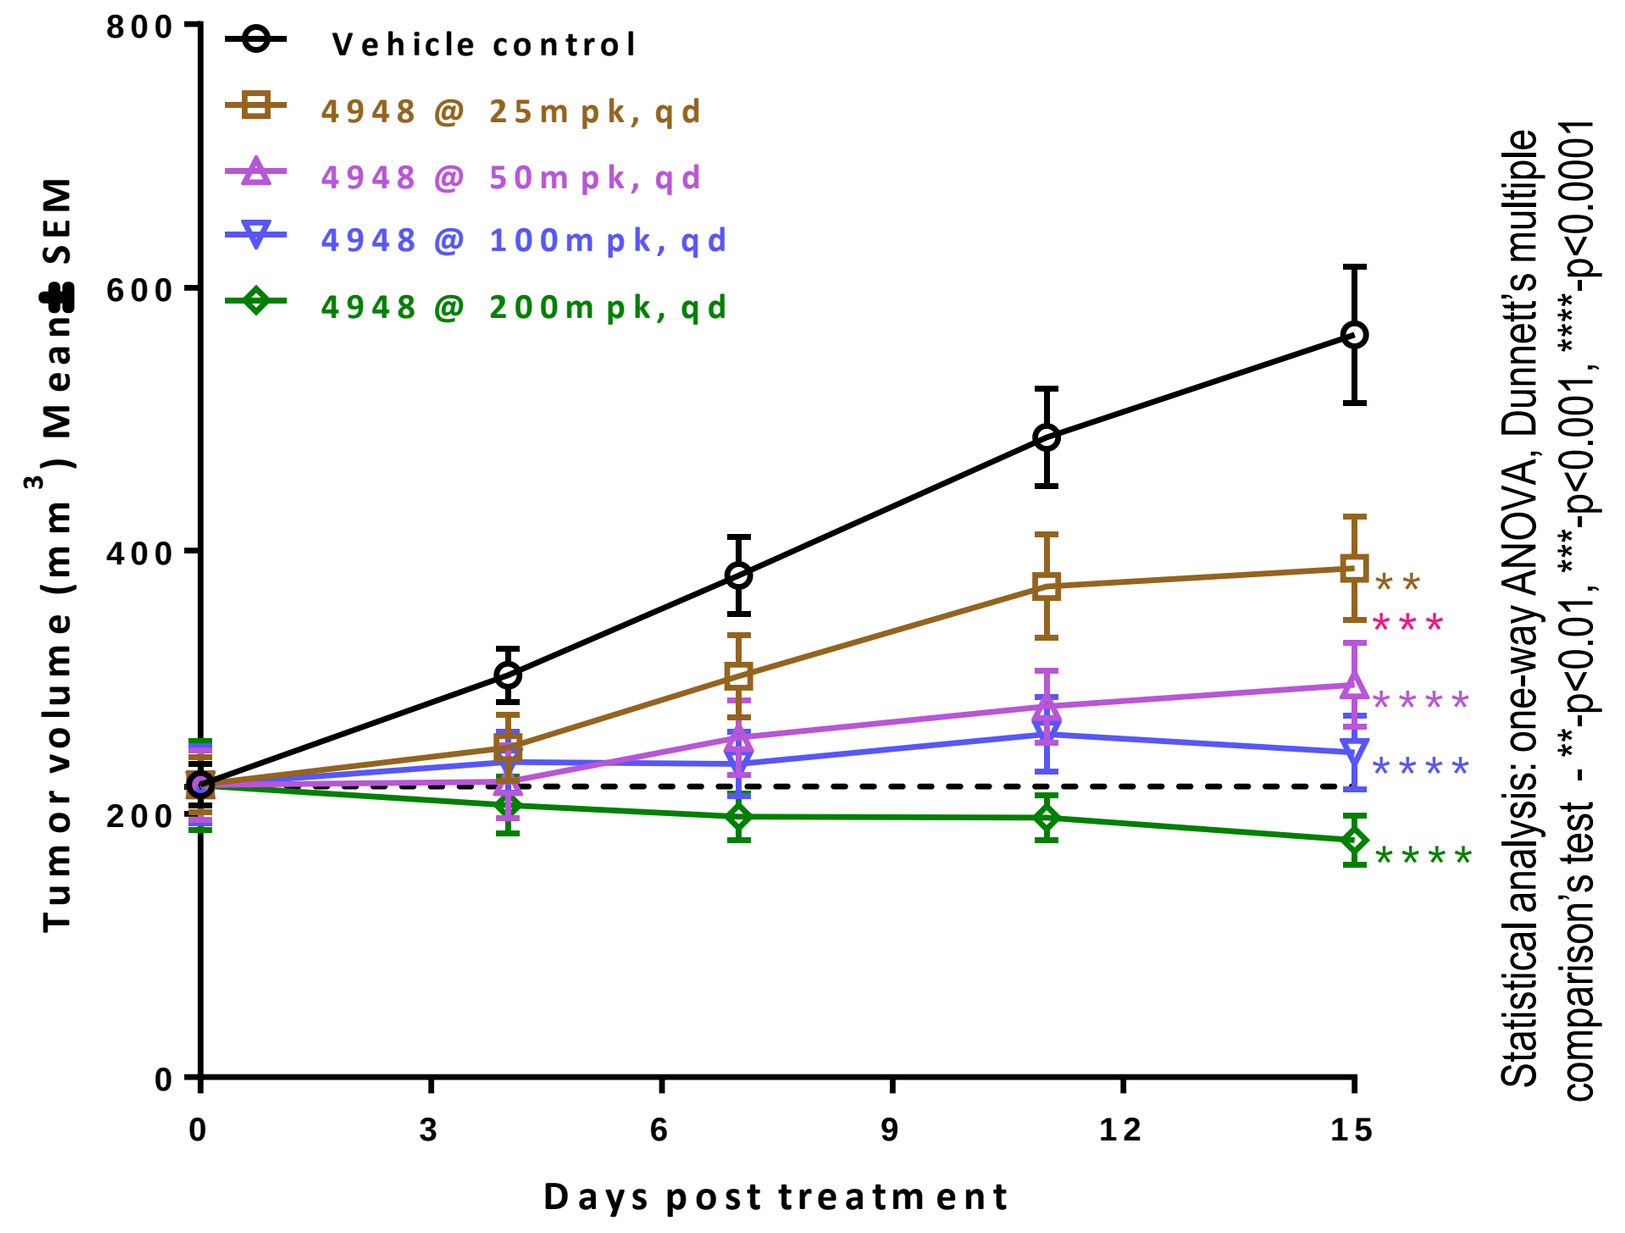
Vehicle control
4948 @ 25mpk, qd
4948 @ 50mpk, qd
4948 @ 100mpk, qd
4948 @ 200mpk, qd
Tumor volume (mm3) Mean ± SEM
800
600
400
200
0
0
3
6
9
12
15
Days post treatment
Statistical analysis: one-way ANOVA, Dunnett's multiple comparison's test - **-p<0.01, ***-p<0.001, ****-p<0.0001

Treatment groups (n=9)	%TGI (Day-15)
AU-4948 @ 25 mpk, po,qd	52**
AU-4948 @ 50 mpk, po,qd	78****
AU-4948 @ 100 mpk, po,qd	93****
AU-4948 @ 200 mpk, po,qd	119****

- AU-4948 treatment led to dose dependent inhibition of tumor growth with MED of 50 mpk
- No body weight reduction in treatment groups
- Well tolerated with no clinical signs or gross pathological changes

PD modulation in OCI-Ly3 tumors by AU-4948 in Single Dose PK/PD Study

AU-4948 treated (50 mpk)	Average % inhibition of p-IRAK1	Average % increase in total IRAK1
0.5 hr	-14	14
1 hr	-50	2
2 hr	-21	-8
4 hr	-19	20
8 hr	31	42

- Significant inhibition of p-IRAK1 observed 8 hours after dosing
- Maximal inhibition of p-IRAK1 likely beyond 8 hours

In-Vitro Toxicity Profile of AU-4948

Parameter	Profile
hERG (patch clamp)	<10% inhibition at 30µM
Ames test	Non-mutagenic in five strains of Salmonella typhimurium (@5 mg/plate)
CYP inhibition (8 isoforms)	IC_{50} >50µM
CYP induction (3 major isoforms)	No CYP induction (tested at 10 µM)
CEREP-44 panel	No Significant inhibition at 10 µM

Clean in-vitro toxicity profile
- No hERG inhibition
- Negative in Ames test
- No CYP inhibition/induction

Preliminary In-Vivo Toxicity Profile of AU-4948

Parameter	Profile
Tolerability in mice MTD study	Well tolerated up to 200 mg/kg
Minimal Efficacious Dose (MED) in OCI-Ly3 model	50mpk (78 % TGI)
Therapeutic Window*	**7.3 fold**

- Excellent therapeutic window based on mice MTD study
- Repeated dose safety evaluation in rodents in progress

*Fold window based on exposure at 200 mpk in MTD study vs efficacious exposure at 50 mpk

Conclusion

- Potent IRAK-4 inhibitors from multiple chemically distinct series identified
- Dose dependent inhibition of TLR4 induced TNFα release demonstrated in-vivo
- Dose dependent inhibition of tumor growth demonstrated in OCI-Ly3 model with MED of 50 mpk
- PD modulation demonstrated in OCI-Ly3 model in a single dose PK/PD study
- AU-4948 was well tolerated at all tested doses in OCI-Ly3 model
- Repeated dose safety evaluation of AU-4948 in rodents in progress

AACR-NCI-EORTC 2015

Contact: murali_r@aurigene.com