MIKIE: A Randomized, Double-Blind, Regimen-Controlled, Phase 2 Study to Assess the Efficacy and Safety of Two Different Vismodegib Regimens in Patients With Multiple Basal Cell Carcinomas

9509

Gary Rogers,[1] Rainer Kunstfeld,[2] Dirk Schadendorf,[3] Axel Hauschild,[4] Ian Maher,[5] David Zloty,[6] Bruno Labeille,[7] Jean-Jacques Grob,[8] Susana Puig,[9] Daniel Bergstrom,[10] Frank Gilberg,[10] Brigitte Dréno[11]

[1]Tufts University School of Medicine, Boston, MA, USA; [2]Medical University of Vienna, Vienna, Austria; [3]Universitätsklinikum Essen, Essen, Germany; [4]University Hospital Schleswig-Holstein, Kiel, Germany; [5]Saint Louis University Medical School, St. Louis, MO, USA; [6]University of British Columbia, Vancouver, BC, Canada; [7]University Hospital of Saint-Etienne, Saint-Etienne, France; [8]Aix-Marseille University, Timone Hospital APHM, Marseille, France; [9]Hospital Clinic de Barcelona, Barcelona, Spain; [10]F. Hoffmann-La Roche, Ltd., Basel, Switzerland; [11]Nantes University, Nantes, France

INTRODUCTION

- Abnormal Hedgehog pathway signaling is the key molecular driver of basal cell carcinoma (BCC) and was first identified in patients with BCC nevus syndrome (BCCNS), also known as Gorlin syndrome[1,2]
- Vismodegib is a first-in-class, oral, selective Hedgehog pathway inhibitor[3] that was approved by the US Food and Drug Administration and the European Medicines Agency for the treatment of adults with metastatic BCC, or with locally advanced BCC inappropriate for surgery or radiotherapy[4,5]
- There is a high unmet need for long-term effective treatments in patients with multiple BCCs
 - Vismodegib has been shown to reduce BCC tumor burden and the rate of appearance of new BCCs in patients with BCCNS; however, the occurrence of chronic, low-grade adverse events (AEs) means that long-term treatment is not tolerable for the majority of patients[6]
- An intermittent dosing regimen might provide a management strategy to help improve tolerability and provide a long-term treatment option
- The MIKIE study (ClinicalTrials.gov ID, NCT01815840) was designed to assess the efficacy and safety of 2 long-term, intermittent vismodegib dosing regimens in patients with multiple BCCs

OBJECTIVES

- **Primary objective:** Assess the relative reduction from baseline (%) in the number of clinically evident BCCs at Week 73 (ie, after 72 weeks of treatment) for the 2 treatment regimens
- **Secondary objective:** Assess, by treatment regimen, the following at Week 73:
 - Relative reduction from baseline (%) in total size of 3 target BCC lesions (largest visible lesions, at least 5 mm in the longest diameter) in individual patients
 - Proportion of patients with at least 50% reduction in the number of BCCs
 - Number of new BCCs
 - Recurrence rate (eg, total number of all BCCs relative to baseline and Week 73) at 12, 24, and 52 weeks after study drug discontinuation
 - Dropout rate and tolerability
 - Patient-reported outcomes, quality of life (Skindex-16 questionnaire)
 - Pharmacokinetic outcomes for selected sites
- **Exploratory objective:** Explore the biomarker profile in patients with multiple BCCs

METHODS

Study Design

- This was a randomized, double-blind, regimen-controlled, phase 2 study of vismodegib in adult patients with multiple BCCs amenable to surgery (including BCCNS)
- Eligible patients were stratified according to BCCNS status, region, and immunosuppression
- Enrolled patients were randomly assigned (1:1) to 1 of 2 treatment arms **(Figure 1)**:
 - **Treatment arm A:** 150 mg vismodegib orally, once daily, continuously for 12 weeks, followed by 8 weeks of placebo. This intermittent schedule was repeated 3 times and followed by a final course of 12 weeks of vismodegib
 - **Treatment arm B:** 150 mg vismodegib orally, once daily, continuously for 24 weeks. This was followed by intermittent treatment of 8 weeks of placebo and 8 weeks of vismodegib. This intermittent schedule was repeated 3 times

Figure 1. Study design.

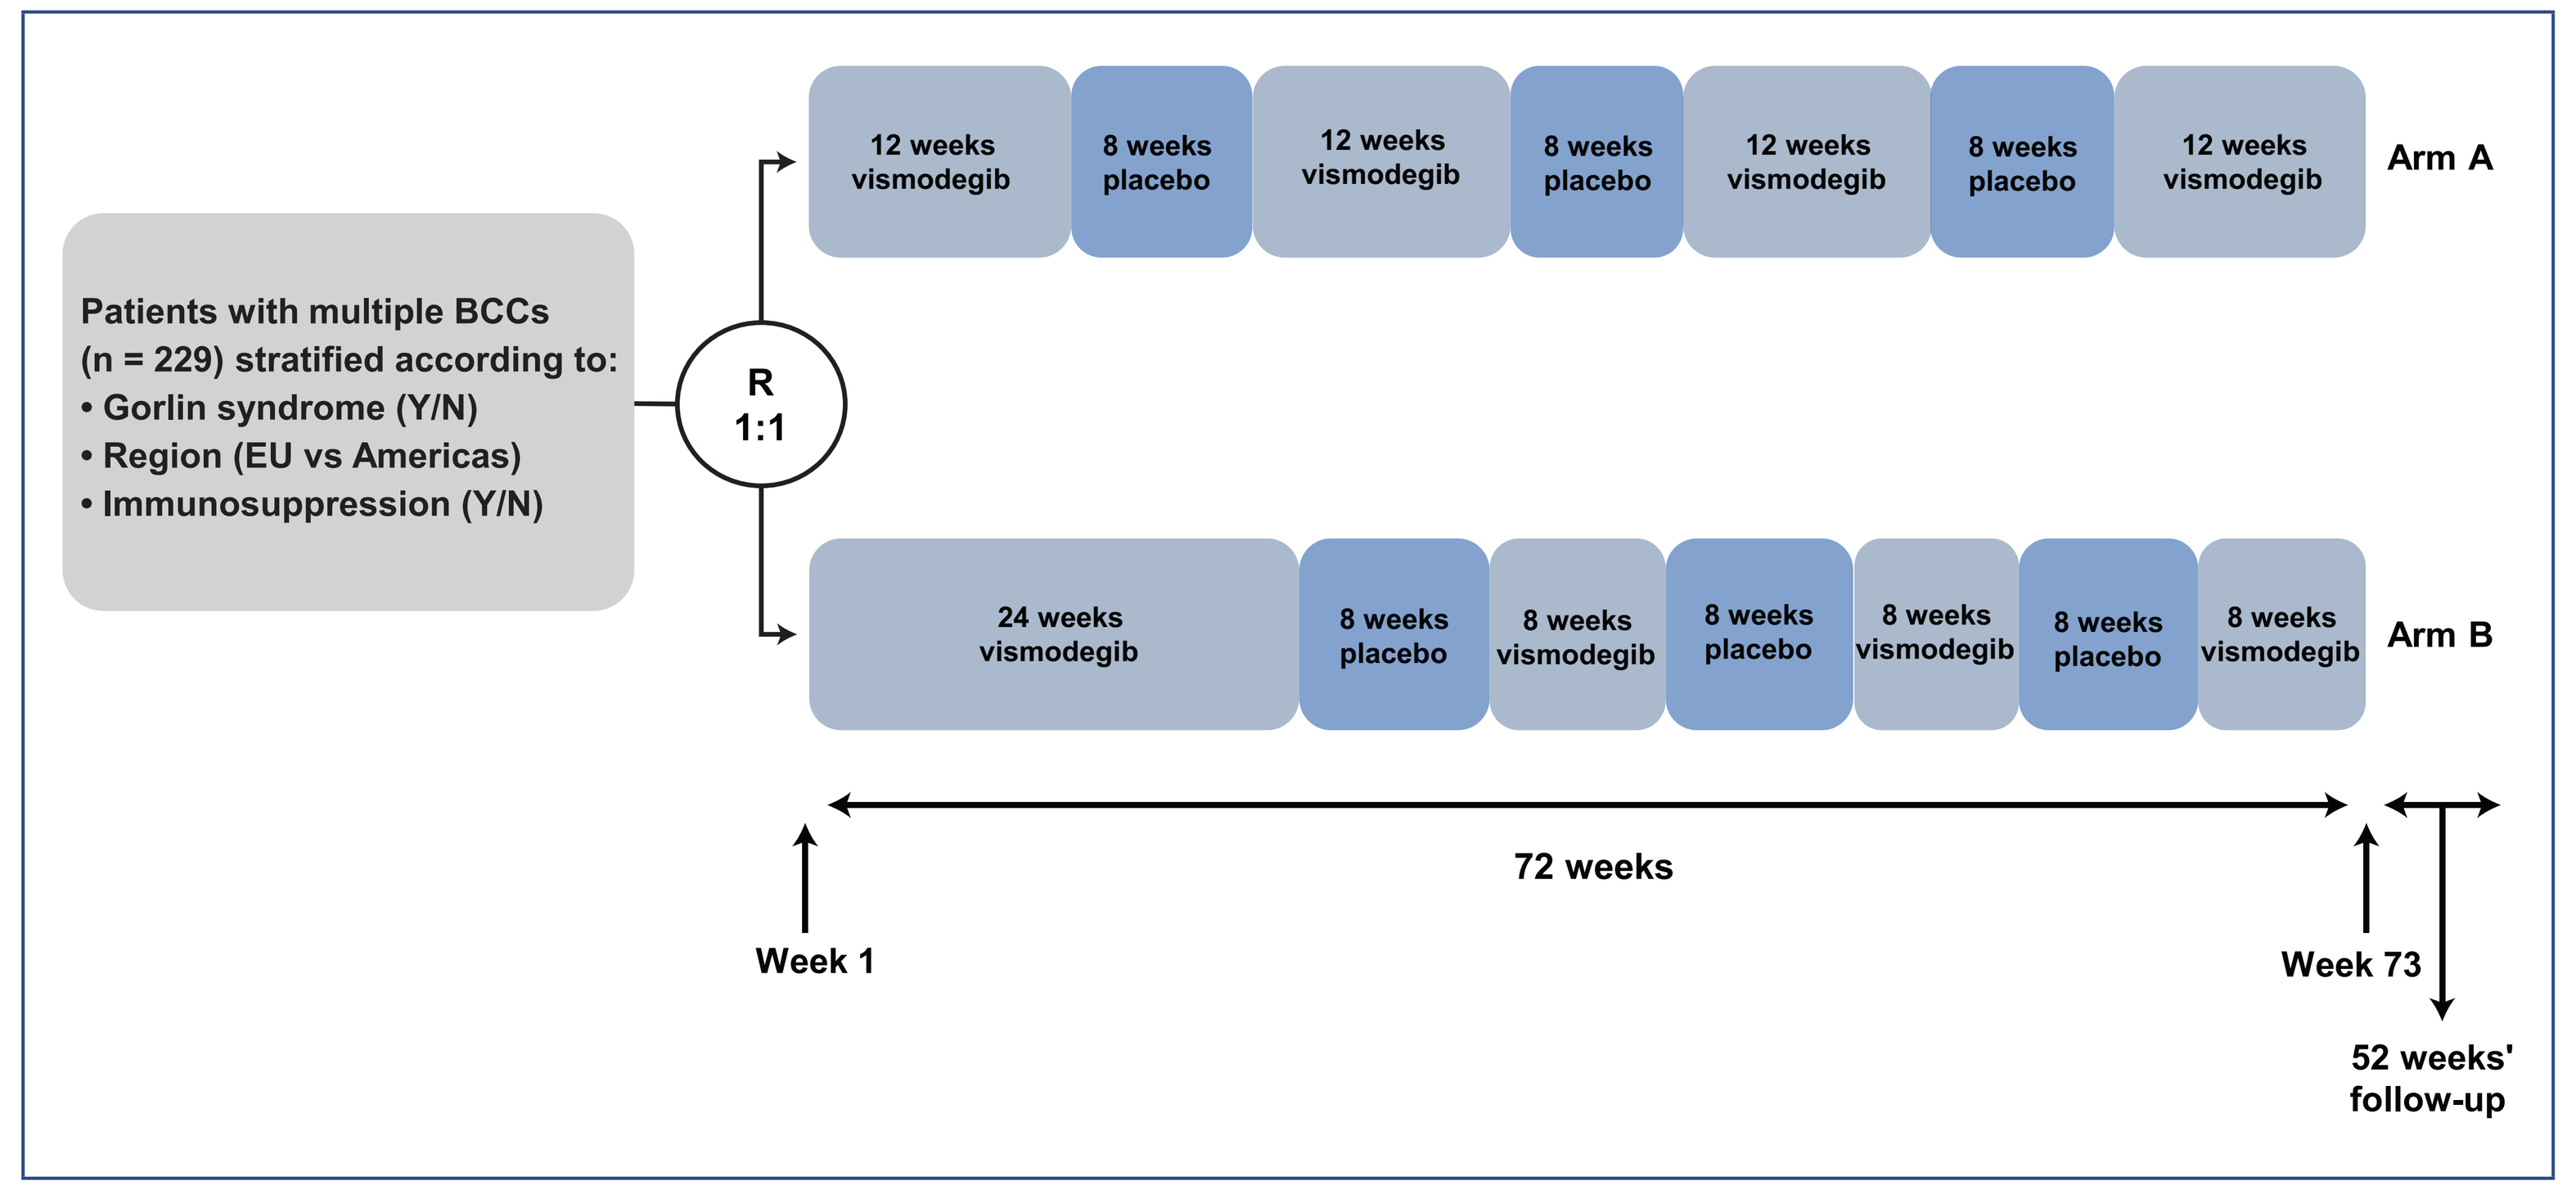
12 weeks vismodegib
8 weeks placebo
12 weeks vismodegib
8 weeks placebo
12 weeks vismodegib
8 weeks placebo
12 weeks vismodegib
Arm A
Patients with multiple BCCs (n = 229) stratified according to:
• Gorlin syndrome (Y/N)
• Region (EU vs Americas)
• Immunosuppression (Y/N)
R 1:1
24 weeks vismodegib
8 weeks placebo
8 weeks vismodegib
8 weeks placebo
8 weeks vismodegib
8 weeks placebo
8 weeks vismodegib
Arm B
72 weeks
Week 1
Week 73
52 weeks' follow-up

BCC, basal cell carcinoma; EU, European Union; R, randomized.

Patient Eligibility

- Key eligibility criteria are summarized in **Table 1**

Table 1. Key Eligibility Criteria

Inclusion criteria	Exclusion criteria
Age ≥18 years	Metastatic BCC
Multiple BCCs, including BCCNS with significant burden of skin disease	Locally advanced BCC lesion considered inoperable or for which surgery is medically contraindicated, including: • BCC that has recurred in the same location after 2 or more surgical procedures and for which curative resection is unlikely • Substantial anticipated morbidity and/or deformity from surgery
6 or more clinically evident BCCs at the time of randomization, at least 3 of which measure ≥5 mm in diameter (target lesions)	Recent (within 28 days) or current participation in another experimental drug study
1 or more target lesions with a histopathologically confirmed diagnosis of BCC (2 mm punch biopsy)	Uncontrolled medical illness
ECOG PS of 0, 1, or 2	

BCC, basal cell carcinoma; BCCNS, BCC nevus syndrome; ECOG PS, Eastern Cooperative Oncology Group performance status.

Statistical Considerations

- The planned sample size of approximately 200 patients was based on the primary end point and 95% confidence intervals (CIs) for each treatment regimen. No power calculation was performed for any other end point
- There was no formal statistical hypothesis for the treatment comparison because the study was not designed to show any statistically significant difference between treatment arms. Each arm was assessed separately; however, 95% CIs and *P* values for the comparison were reported for all relevant estimates
- For the primary efficacy analysis, the mean difference in the relative reduction between treatment arms, along with the corresponding 95% two-sided CIs, was estimated by fitting an analysis of covariance (ANCOVA) model, adjusting for stratification factors

RESULTS

Patient Characteristics

- Between April 30, 2013, and April 9, 2014, 263 patients were screened; a total of 229 patients were randomly assigned (116 in treatment arm A and 113 in treatment arm B) at 52 study sites in 10 countries (intention-to-treat [ITT] analysis population)
 - Of these, 227 received at least 1 dose of vismodegib (safety analysis population)
- Baseline characteristics, demographics, and stratification factors are summarized in **Table 2**

Table 2. Baseline Characteristics, Demographics, and Stratification Factors

Characteristic	Treatment arm A n = 116	Treatment arm B n = 113	Total N = 229
Sex, n (%)			
Men	81 (69.8)	88 (77.9)	169 (73.8)
Women	35 (30.2)	25 (22.1)	60 (26.2)
Age, y, mean (range)	61 (27-89)	60 (27-91)	61 (27-91)
<65 y, n (%)	63 (54.3)	64 (56.6)	127 (55.5)
≥65 y, n (%)	53 (45.7)	49 (43.4)	102 (44.5)
Confirmed diagnosis of BCCNS, n (%)			
Yes	44 (37.9)	41 (36.3)	85 (37.1)
No	72 (62.1)	72 (63.7)	144 (62.9)
Geographic region, n (%)			
America	36 (31.0)	35 (31.0)	71 (31.0)
Europe	80 (69.0)	78 (69.0)	158 (69.0)
Immunosuppression status, n (%)			
Immunocompetent	116 (100.0)	112 (99.1)	228 (99.6)
Immunosuppressed	0	1 (0.9)	1 (0.4)
Baseline ECOG PS, n (%)			
0	97 (88.2)	93 (83.0)	190 (85.6)
1	12 (10.9)	14 (12.5)	26 (11.7)
2	1 (0.9)	5 (4.5)	6 (2.7)
Procedure history related to BCC, n (%)			
Yes	105 (90.5)	90 (79.6)	195 (85.2)
Procedure type, n (%)			
Complex surgical excision	16 (13.9)	5 (4.4)	21 (9.2)
Cryotherapy	12 (10.4)	8 (7.1)	20 (8.8)
Mohs surgery	25 (21.7)	24 (21.2)	49 (21.5)
Other	23 (20.0)	25 (22.1)	48 (21.1)
Simple surgical excision	78 (67.8)	67 (59.3)	145 (63.6)

BCC, basal cell carcinoma; BCCNS, BCC nevus syndrome, ECOG PS, Eastern Cooperative Oncology Group performance status.

Patient Disposition

- In the ITT analysis population (n = 229), treatment was discontinued in 108 patients (47.2%) because of the following: AEs (53 patients [23.1%]); withdrawal of consent (26 patients [11.4%]); patient refused treatment (10 patients [4.4%]); investigator's decision (8 patients [3.5%]); disease progression (6 patients [2.6%]); administrative/other (2 patients [0.9%]); lost to follow-up (1 patient [0.4%]); major protocol deviation (1 patient [0.4%]); sponsor terminated treatment (1 patient [0.4%])
- In the safety analysis population (n = 227), the mean overall treatment duration was 11.4 months (11.8 months for treatment arm A and 11.0 months for treatment arm B)
- Overall, the majority of patients were treatment adherent: 97.4% (n = 111) in treatment arm A and 94.7% (n = 107) in treatment arm B

Efficacy

- The primary efficacy analysis was performed for the ITT analysis population (n = 229)
- The highest mean relative reduction from baseline in number of clinically evident BCCs was observed at end of treatment (EOT) and was higher in treatment arm A (62.7%; 95% CI, 53.0-72.3) than in treatment arm B (54.0%; 95% CI, 43.6-64.4)
 - For the ANCOVA models, the treatment arm *P* values were similar for model A (adjusting for stratification factors) and model B (excluding stratification factors); the values were not statistically significant (*P* = 0.2132 and *P* = 0.2443 for model A and model B, respectively)
 - A percentage relative reduction from baseline in the number of clinically evident BCCs was reported from Cycle 3 (Week 9) in treatment arm A (19.1%; 95% CI, 14.4-23.8) and treatment arm B (16.9%; 95% CI, 11.1-22.6)
 - A marked increase in relative reduction from baseline was observed at every tumor evaluation until Cycle 9
 - Mean percentage relative reduction of clinically evident BCCs over time is shown in **Figure 2**

Figure 2. Mean percentage relative reduction in the number of clinically evident BCCs.

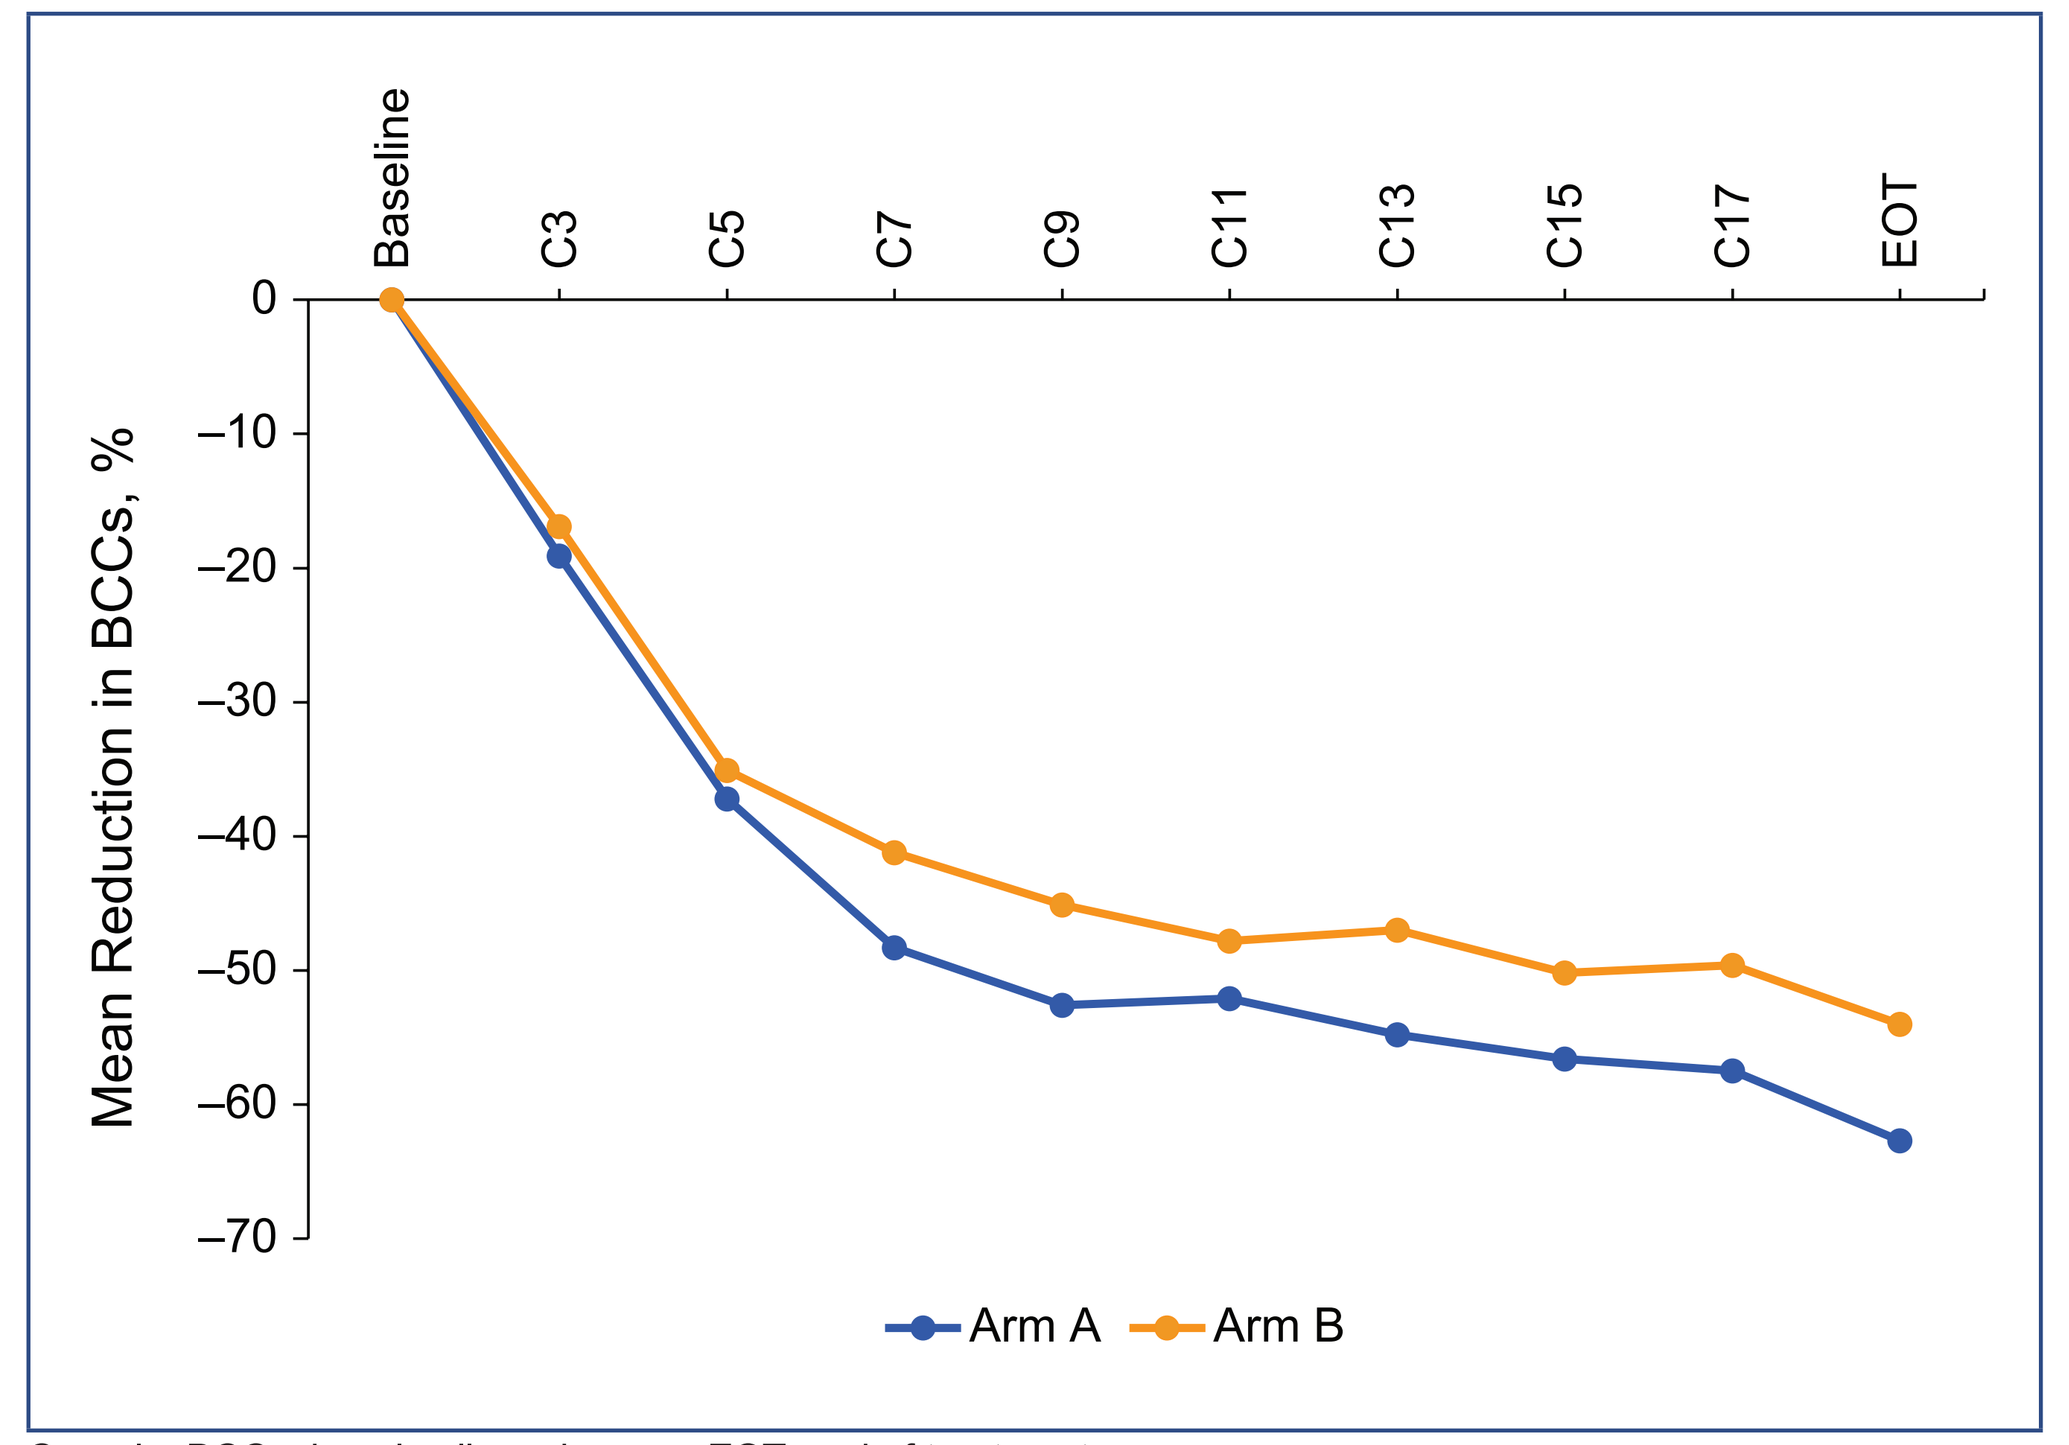
Baseline
C3
C5
C7
C9
C11
C13
C15
C17
EOT
0
–10
–20
–30
–40
–50
–60
–70
Mean Reduction in BCCs, %
Arm A
Arm B

C, cycle; BCCs, basal cell carcinomas; EOT, end of treatment.

- A greater mean percentage relative reduction in the total size of 3 target BCC lesions was observed in treatment arm A (82.9%; 95% CI, 77.4-88.4) than in treatment arm B (68.8%; 95% CI, 57.4-80.2) at EOT
 - The difference in the mean relative reduction between treatment arms was –15.2% (95% CI, –27.4 to –3.0) from the ANCOVA model (*P* = 0.0146)
- Many patients in both treatment arms experienced a reduction in total number of BCCs of ≥50%: 65.5% (n = 76) in treatment arm A and 50.4% (n = 57) in treatment arm B at EOT
- The number of new BCC lesions observed at EOT, compared with the number observed at baseline, was similar between treatment arms
- The majority of patients did not have new lesions: 76.6% (n = 72; 95% CI, 66.7-84.7) in treatment arm A and 74.4% (n = 64; 95% CI, 63.9-83.2) in treatment arm B
- **Table 3** summarizes the primary and secondary efficacy outcomes

Table 3. Primary and Secondary Efficacy Outcomes

	Treatment arm A n = 116	Treatment arm B n = 113	Difference (arm A to arm B) [95% CI][a]	Treatment arm *P*-value (from ANCOVA)[a]	
				Model A[b]	Model B[c]
Mean relative reduction from baseline in total number of BCC lesions at EOT, %	62.7	54.0	–8.9% [–23.0 to 5.2]	0.2132	0.2443
Relative reduction in total size of 3 target BCCs, %	82.9	68.8	–15.2 [–27.4 to –3.0]	0.0146	
Proportion of patients with at least 50% reduction in total number of BCCs from baseline at EOT, n (%)	76 (65.5)	57 (50.4)	–15.1% [–27.7 to –2.4]	< 0.05	
Number of patients without new BCC lesions at EOT compared with baseline, n (%)	72 (76.6)	64 (74.4)	–2.2% [–14.8 to 10.4]	> 0.05	

ANCOVA, analysis of covariance; BCC, basal cell carcinoma; CI, confidence interval; EOT, end of treatment.
[a]Exploratory analysis suggested by ethics committee. The study was not designed for a statistical comparison between treatment arms.
[b]Model A: ANCOVA model adjusted for stratification factors.
[c]Model B: ANCOVA model not adjusted for stratification factors.

Safety

- The safety results are derived from the safety analysis population (all ITT analysis-population patients who received at least 1 dose of the study treatment) (n = 227)
- The majority of patients (223 [98.2%]) experienced at least 1 treatment-emergent adverse event (TEAE) during the course of the study: 113 patients (99.1%) in treatment arm A and 110 patients (97.3%) in treatment arm B
- The most common TEAEs, occurring in ≥10% of patients, are listed in **Table 4**
- Grade ≥3 TEAEs were reported by 70 patients (30.8%): 31 patients (27.2%) in treatment arm A and 39 patients (34.5%) in treatment arm B
 - The most frequently reported grade ≥3 TEAEs were muscle spasm (16 [7.0%]), anemia (4 [1.8%]), dysgeusia (3 [1.3%]), pneumonia (3 [1.3%]), and hypophosphatemia (3 [1.3%])
- 41 patients (18.1%) experienced a serious TEAE. The number of patients was similar between the 2 treatment arms: 22 patients (19.3%) in treatment arm A and 19 patients (16.8%) in treatment arm B
 - 8 patients (3.5%) experienced serious TEAEs related to study treatment: hepatic enzyme increase (1 [0.4%]), platelet count decrease (1 [0.4%]), pseudolymphoma (1 [0.4%]), pancreatitis acute (1 [0.4%]), asthenia (1 [0.4%]), dehydration (1 [0.4%]), arthralgia (1 [0.4%]), lethargy (1 [0.4%]), and pulmonary embolism (1 [0.4%])
- At data cutoff, 4 patients (1.8%) had died as a result of TEAEs while on study or during follow-up: 2 patients (1.8%) in treatment arm A and 2 patients (1.8%) in treatment arm B
 - The TEAEs that led to death were pulmonary embolism (2 patients [0.9%]: 1 patient [0.9%] each in treatment arm A and treatment arm B, respectively), cardiogenic shock (1 patient [0.9%] in treatment arm B), and pneumonia (1 patient [0.9%] in treatment arm A)
 - Of these TEAEs, only the pulmonary embolism in treatment arm A was suspected of being related to study treatment by the study investigator. Other possible etiologic factors for the event included reduced activity after surgical removal of a congenital benign cyst in the third ventricle that was detected on study Day 177
- 53 patients (23.3%) experienced a TEAE that led to discontinuation of study treatment. The number of patients was similar between the 2 treatment arms (23 patients [20.2%] in treatment arm A, 30 patients [26.5%] in treatment arm B)
 - The most frequently reported TEAEs that led to discontinuation of study treatment were muscle spasm (21 patients [9.3%]) and dysgeusia (13 patients [5.7%])
 - 45 patients (19.8%) experienced a TEAE that led to an interruption of study treatment (24 patients [21.1%] in treatment arm A and 21 patients [18.6%] in treatment arm B)
 - The most frequently reported TEAE that led to an interruption of study treatment was muscle spasm (9 patients [4.0%])

Table 4. Most Common TEAEs, Occurring in ≥10% of Patients

	Patients with TEAEs, n (%)		
	Treatment arm A n = 114	Treatment arm B n = 113	Total N = 227
Any TEAE	113 (99.1)	110 (97.3)	223 (98.2)
Muscle spasm	83 (72.8)	93 (82.3)	176 (77.5)
Dysgeusia	75 (65.8)	75 (66.4)	150 (66.1)
Alopecia	72 (63.2)	73 (64.6)	145 (63.9)
Fatigue	24 (21.1)	26 (23.0)	50 (22.0)
Weight decreased	24 (21.1)	21 (18.6)	45 (19.8)
Decreased appetite	21 (18.4)	17 (15.0)	38 (16.7)
Diarrhea	20 (17.5)	18 (15.9)	38 (16.7)
Nausea	23 (20.2)	14 (12.4)	37 (16.3)
Asthenia	15 (13.2)	20 (17.7)	35 (15.4)
Arthralgia	18 (15.8)	16 (14.2)	34 (15.0)
Myalgia	18 (15.8)	12 (10.6)	30 (13.2)
Ageusia	14 (12.3)	13 (11.5)	27 (11.9)
Blood creatine phosphokinase level increase	11 (9.6)	15 (13.3)	26 (11.5)
Headache	11 (9.6)	12 (10.6)	23 (10.1)

TEAE, treatment-emergent adverse event.

CONCLUSIONS

- Both treatment regimens were effective, and planned treatment breaks did not appear to affect effectiveness in either regimen
 - Treatment arm A was associated with numerically better outcomes in terms of efficacy and discontinuation rate
- The safety profiles of both regimens were similar and were consistent with previous clinical experience[7,8]
- Intermittent dosing schedules might allow patients with multiple BCCs to derive benefit from long-term vismodegib treatment

REFERENCES

1. Scales SJ, de Sauvage FJ. *Trends Pharmacol Sci.* 2009;30:303-312.
2. Epstein EH. *Nat Rev Cancer.* 2008;8:743-754.
3. Basset-Seguin N et al. *Mol Cancer Ther.* 2015;14:633-641.
4. Erivedge [prescribing information]. South San Francisco, CA: Genentech, Inc., May 2015.
5. Erivedge (vismodegib) [summary of product characteristics]. Welwyn Garden City, UK: Roche Products Ltd.; February 2016.
6. Tang JY et al. *N Engl J Med.* 2012;366:2180-2188.
7. Sekulic A et al. *N Engl J Med.* 2012;366:2171-2179.
8. Hansson J et al. Presented at: 2016 American Society of Clinical Oncology Annual Meeting; June 3-7, 2016; Chicago, IL. Abstract.

ACKNOWLEDGMENTS

The authors thank the participating investigators, patients, and their families, and current and past members of the Roche and Genentech vismodegib teams. This analysis was funded by F. Hoffman-La Roche, Ltd. Third-party medical writing support was provided by Clare Driscoll, PhD (ApotheCom, San Francisco, CA), and was funded by F. Hoffman-La Roche, Ltd.

Roche is developing vismodegib under a collaboration agreement with Curis, Inc. Vismodegib was discovered by Genentech and jointly validated by Genentech and Curis through a series of preclinical studies. Through this collaboration, Genentech (USA), Roche (outside the USA, excluding Japan and Korea), and Chugai Pharmaceuticals (Japan) are responsible for the clinical development and commercialization of vismodegib.

Contact Dr. Gary Rogers at gary.rogers@tufts.edu for questions or comments.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

http://bit.ly/1NWSt11

Presented at the 2016 American Society of Clinical Oncology Annual Meeting; June 3-7, 2016, Chicago, IL, USA